Development of a multiparametric cell imaging assay to assess inverse regulation of cancer associated biomarkers

T. Jones[1], D. Kostomiris[2], M. Bilyard[1], A. Ahmed[3], A. Madin[4], G. Kauffman[5], S. Mlynarski[5], R. Ziegler[5], H. Liu[5], N. Umbreit[5] & D. Zindel[1]

AstraZeneca Mechanistic and Structural Biology, Cambridge, UK[1], AstraZeneca Mechanistic and Structural Biology, Waltham, United States[2], AstraZeneca Cellular Assay Development, Cambridge, UK[3], AstraZeneca Hit Discovery, Cambridge, UK[4] & AstraZeneca Oncology, Waltham, United States[5]

Abstract

Background: Our protein of interest (POI) is overexpressed in cancers, associates with a poor prognosis, and is a potential therapeutic target
Objective: To develop a multiparametric cellular high content screening (HCS) assay that allows co-investigation of POI proximal and distal effects following small molecule inhibitor treatment
Methods: Co-assessment of distal biomarker upregulation and POI expression was performed by immunofluorescence using the MCF-7 cell line, 28h post-treatment with a literature tool inhibitor
Results: We observe concentration-dependent inverse biomarker regulation, observing distal biomarker upregulation and POI downregulation in cells treated with literature tool inhibitor
Conclusions: Using a literature tool inhibitor we demonstrate that our HCS assay is suitable to generate cellular SAR-enabling data

Introduction

- Our POI is demonstrated to be overexpressed in a range of cancers, with an established link to a poorer prognosis in solid tumours
- The POI regulates elements of the DNA damage response. Inhibitors of the POI drive an antiproliferative response *in vitro* but have not been progressed or validated clinically
- Here the objective was to develop a multiparametric cellular HCS assay that permits the co-assessment of POI proximal and POI distal biomarkers to support our small molecule inhibitor project

Methods

- MCF-7 cells were seeded into 384-well plates overnight before compound treatment. 12 point, 0.5 log interval response curves were generated from 30 µM for AC50 estimates
- Cells were fixed with 4% paraformaldehyde and permeabilized and stained in 1.1% BSA, 0.1% Triton X-100 in PBS. Cells were stained overnight at 4°C with primary antibodies and 1h at room temperature for secondary antibodies with Hoechst stain. Plate washing was performed with the Biotek EL406 (Agilent) and Blue®washer (BlueCatBio)
- Co-assessment of nuclear distal biomarker upregulation (Ch3 - AF647), nuclear POI expression (Ch2 - AF488) and total cell number (Ch1 - Hoechst) was performed using the Cell Insight™ CX5 High Content Screening Platform (Thermo)

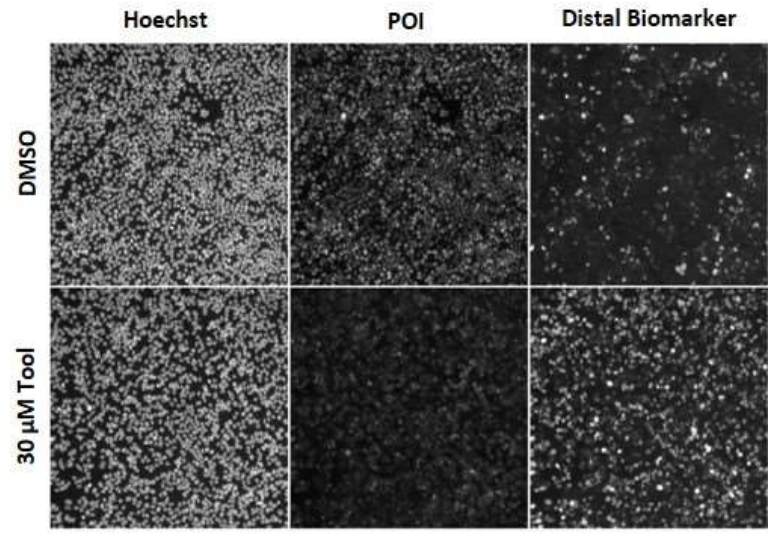
Hoechst
POI
Distal Biomarker
DMSO
30 µM Tool

Figure 1. Representative images of MCF-7 cells fixed 28h post-treatment with vehicle or literature tool inhibitor

Results

Literature tool inhibitor drives concentration-dependent inverse biomarker regulation

In cells 28h post-treatment, literature tool inhibitor is demonstrated to inversely regulate nuclear expression of the distal biomarker and POI

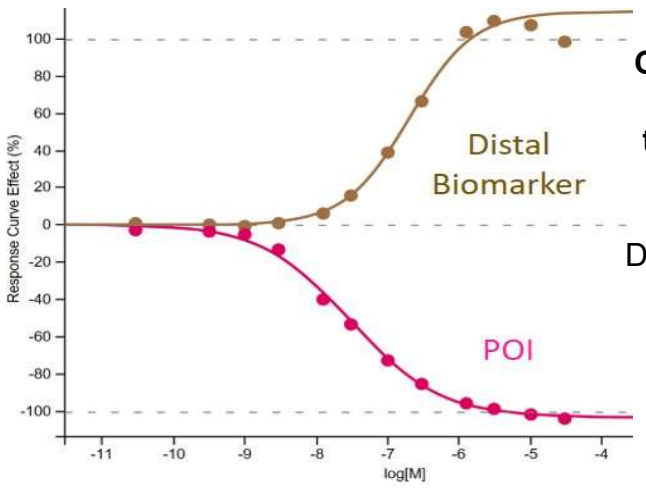
Distal Biomarker
POI
Response Curve Effect (%)
log[M]

Figure 2. Concentration response for tool 28h post treatment:

Distal Biomarker AC50: 0.21 µM

POI AC50: 0.03 µM

Cell number and timecourse data support optimal co-assessment window

Cell number optimization shows 10,000 cells per well provided superior assay performance for assessment of distal biomarker upregulation using an extended compound validation set

	Cells per well		
Assay Parameter	**10,000**	**5,000**	**2500**
RZ'	0.77	0.62	0.40
S/B	2.6	2.4	2.4
Sensitivity*	10/19	6/14	6/14

Table 1. Distal biomarker parameters following 28h compound treatment. (*) Compounds with AC50 < 30 µM

Timecourse optimization shows compound treatment period of 24h provides superior assay parameters to shorter treatments for assessment of POI and distal biomarker co-regulation, using an extended compound validation set

		Timepoint				
Biomarker	**Assay Parameter**	**1h**	**2h**	**4h**	**7h**	**24h**
POI	RZ'	0.66	0.70	0.82	0.82	0.85
	S/B	2.0	2.8	3.1	3.9	3.7
	Sensitivity*	3/20	3/20	5/20	5/20	8/20
Distal Biomarker	RZ'	-9.35	-0.79	0.48	0.41	0.79
	S/B	1.0	1.2	1.7	1.7	3.1
	Sensitivity*	-	-	2/20	2/20	3/20

Table 2. Assay parameters following varying compound treatment time. (*) Compounds with AC50 < 30 µM

Fluorophore swap confirms channel specificity

Switch of POI detection fluorophore AF488 to AF647, and the converse for distal biomarker did not impact assay parameters nor compound potency

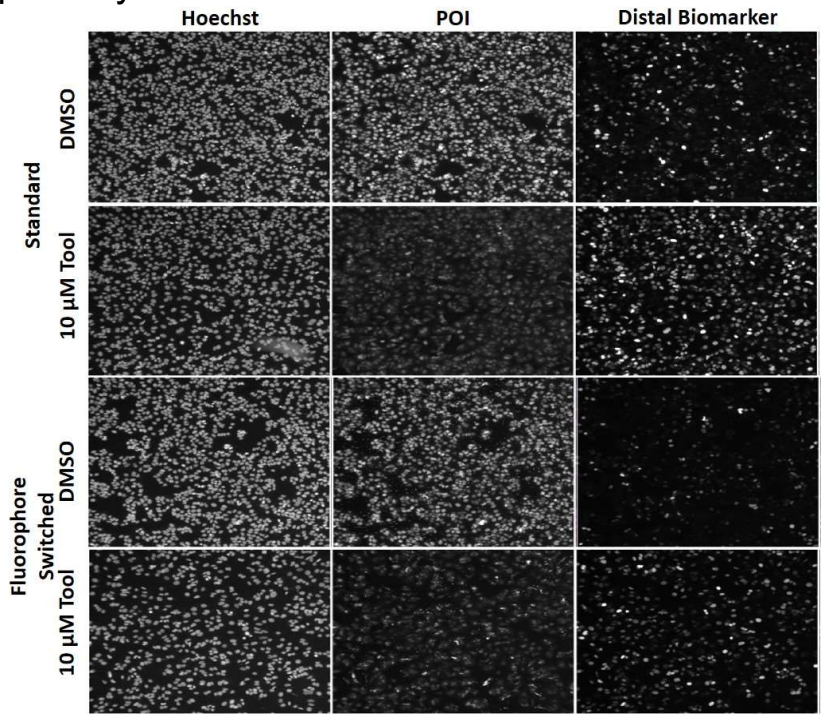
Hoechst
POI
Distal Biomarker
Standard
DMSO
10 µM Tool
Fluorophore Switched
DMSO
10 µM Tool

Figure 3. Representative images of MCF-7 cells stained by standard (top two rows) or fluorophore switched (bottom two rows) secondary antibodies

A dynamic potency range of POI downregulation is detected and complements biochemical data for POI engagement

Using an extended compound validation set, including literature tool inhibitor, we observe robust POI downregulation across a range of potencies 28h post-treatment

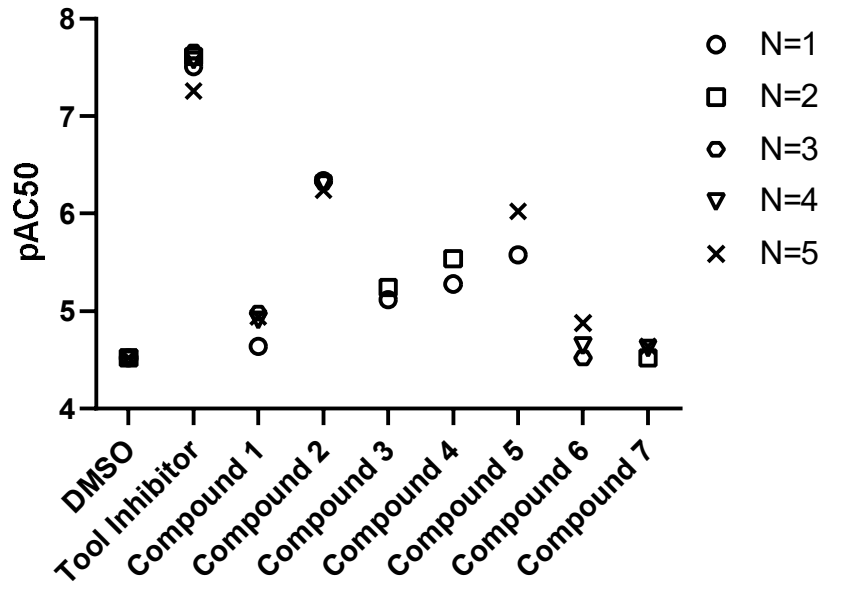
pAC50
N=1
N=2
N=3
N=4
N=5
DMSO
Tool Inhibitor
Compound 1
Compound 2
Compound 3
Compound 4
Compound 5
Compound 6
Compound 7

Figure 4. POI downregulation potencies for tool inhibitor and a range of internal equity 28h post-treatment

We have identified internal equity potentially co-modulating POI and distal biomarker expression out of lead generation campaigns, complementing biochemical binding and catalytic assay data

Readout	Tool Inhibitor	Series 1	Series 2	Series 3	Series 4
Distal Biomarker Cell Upregulation (µM)	0.21	30 (weakly active)	26.83	24.67	23.16
POI Cell Downregulation (µM)	0.03	3.90	5.72	11.87	21.92
POI Biochemical Binding (µM)	0.07	1.21	2.03	3.54	>100
POI Biochemical Catalytic (µM)	0.01	0.66	1.41	2.20	38.97

Table 3. AC50 data for cellular distal biomarker upregulation, cellular POI downregulation, POI biochemical binding and POI biochemical catalytic assays for example chemical series following lead generation

Conclusions

- We have developed a novel, multiparametric cell imaging assay to assess compound induced inverse co-regulation of both POI and distal biomarker expression
- Using a literature tool inhibitor, we can demonstrate that our HCS assay is suitable to generate cellular SAR-enabling data and have identified compounds co-modulating the POI and a distal biomarker in MCF-7 cells
- Next challenges for the team include:
 - To identify additional proximal biomarkers to further support mode of action understanding
 - To identify cellular models and/or assay formats to better discriminate POI specific vs POI non-specific anti-proliferative effects

Acknowledgements
POI biochemical catalytic assay data and binding data were supported by D.K. and M.B, respectively. Distal biomarker assay development was supported by A.A. and D.Z. respectively.

Supported by AstraZeneca

Presented at the ELRIG UK Drug Discovery 2022, Excel London, 04 October 2022