Primary rodent hepatocyte isolation and culture with GMP and research-grade enzyme blends

David Whaley[1], Kimia Damyar[1], Colleen Luong[1], Ivana Xu[1], Gabriel Peixoto[3], Johanna Mönch[4], Michael Alexander[1], Rafal P. Witek[3], Jonathan RT Lakey[1,2] , Tom Bates[5]

1 Department of Surgery, University of California Irvine, 333 City Blvd West, Suite 1600, Orange, CA 92868
2 Department of Biomedical Engineering, University of California Irvine, 402 E Peltason Dr, Irvine, CA 92617
3 Ambys Medicines, 131 Oyster Point Blvd. Suite 200, South San Francisco, CA 94080
4 Nordmark Pharma GmbH Uetersen, Germany
5 AMS Biotechnology (Europe) Ltd, United Kingdom

School of Medicine
University of California · Irvine
Discover · Teach · Heal

INTRODUCTION

Hepatocyte transplantations for acute liver failure and metabolic liver disorders represent a potential alternative to traditional orthotopic liver transplantation (OLT). Cultured primary hepatocytes are an important tool for *in vitro* pharmacological studies. The isolation of primary hepatocytes for clinical applications must comply with current Good Manufacturing Practices (cGMP) regulations, which may include the use of digestion enzymes produced to cGMP standards. In this study, we compare VitaCyte® GMP-grade Collagenase MA in combination with BP Protease to Nordmark® Collagenase NB 6 GMP Grade. Additionally, we compare non-GMP-grade Nordmark® Collagenase NB 4G Proved Grade to GMP-grade Nordmark® Collagenase NB 6 to identify a high-quality, cost-effective alternative for hepatocyte isolation in non-clinical research applications. Advances in clinical hepatocyte transplantation and drug studies depend on the development of standardized isolation and culturing protocols, as well as high-quality enzymes for liver tissue digestion.

MATERIALS & METHODS

<u>Enzyme reconstitution & perfusion</u>

- All animal procedures performed under the approved University of California, Irvine Institutional Animal Care and Use Committee (Protocol #: AUP-17-241)
- Hepatocytes obtained from the livers of male Sprague Dawley rats (300-350 g)
- Whole organ *in situ* digestions performed with a GMP-grade VitaCyte® Collagenase MA and BP Protease blend, GMP-Grade Nordmark® Collagenase NB 6 and non-GMP-Grade Nordmark® Collagenase NB 4G
- Reconstitution of lyophilized enzymes with Ca^{2+}-free Hanks' Balanced Salt solution®
- Enzyme concentrations: VitaCyte® Collagenase MA (2500 CDA U/mL; equivalent to 2.5 PZ U/mL) + BP Protease (550 NP U/mL), Nordmark® Collagenase NB 6 (0.125 PZ U/mL), and Nordmark® Collagenase NB 4G (0.125 PZ U/mL)
- Perfusion of livers using modified version of Berry and Friend's two-step method *in situ* via the hepatic portal vein with pre-warmed VitroPrep® Ca^{2+}-free Liver Perfusion Solution I followed by perfusion with pre-warmed VitroPrep® Liver Perfusion Solution II incl. collagenases **(Fig 1)**
- Removal of digested livers + dispersion into pre-warmed isolation media to isolate and purify hepatocytes
- Isolation media: DMEM supplemented with 10% fetal bovine serum (FBS), 1% Insulin Transferrin Selenium (ITS-G), 1% Penicillin-Streptomycin, 10 mM Dexamethasone

<u>Processing & purification</u>

- Straining of digested livers through 250 and 80 µm nylon mesh
- Addition of fresh pre-warmed isolation media to the resulting suspension
- Assessment of pre-purification yield and viability via trypan blue (minimum allowable viability for inclusion: 70%) (**Fig 3, 4**)
- Centrifugation of cell suspensions at 100xg for 10 min + aspiration of supernatant + gentle re-suspension in warm media
- Purification: Isotonic Percoll® gradient, followed by centrifugation at 100xg/10 min
- Aspiration of supernatants + re-suspension of cell pellets in warm media followed by a trypan blue exclusion assay for yield and viability assessment post-Percoll® (**Fig 3, 4**).

<u>Cell culture</u>

- Aliquot of freshly-isolated hepatocytes taken from final purified cell suspension; dilution to final concentration of $1x10^6$ cells/mL with pre-warmed William's-based hepatocyte plating media (10% FBS, 1% GlutaMAX™, 1% Penicillin-Streptomycin, 1% ITS-G, 10 mM dexamethasone)
- Seeding of fresh cells in triplicate on 24-well type 1 collagen-coated plates (density: $2.11x10^5$ cells/cm^2; incubation at 37ºC in 5% CO_2 environment for 4 h)
- Aspiration of plating media; rinsing of each well 1x with 500 µL William's-based maintenance media (1% GlutaMAX™, 1% Penicillin-Streptomycin, 1% ITS-G, 10 mM dexamethasone)
- Addition of 500 µL of Geltrex®-supplemented maintenance media + incubation at 37 ºC in 5% CO_2 for 72 h
- Change of media every 24 h for three days (72 h total) + collected for albumin assay at 72-h time point. Photos taken at 24-h intervals to aid assessing cell polarization and morphology **(Fig 2)**

<u>Cryopreservation</u>

- Cryopreservation of non-cultured hepatocytes using standard method
- Centrifugation of 180 million viable freshly-isolated hepatocytes at 100 x g for 5 min
- Aspiration of supernatant + re-suspension of cell pellet in VitroPrep® NG5A CryoPreserv cryopreservation media (final concentration:10^7 viable cells/mL)
- Cooling of samples to -90 + placing in vapor-phase LN_2 at -190 ºC

<u>Cell Validation with Albumin ELISA</u>

- Hepatocyte confirmation through ELISA assay via Abcam® Rat Albumin ELISA Kit (**Fig 5**)
- Collection of culture media in triplicate + pooling after 72 h
- Assay: Fresh hepatocyte media + pooled media from each group + protein standard in duplicate per manufacturer protocol

RESULTS

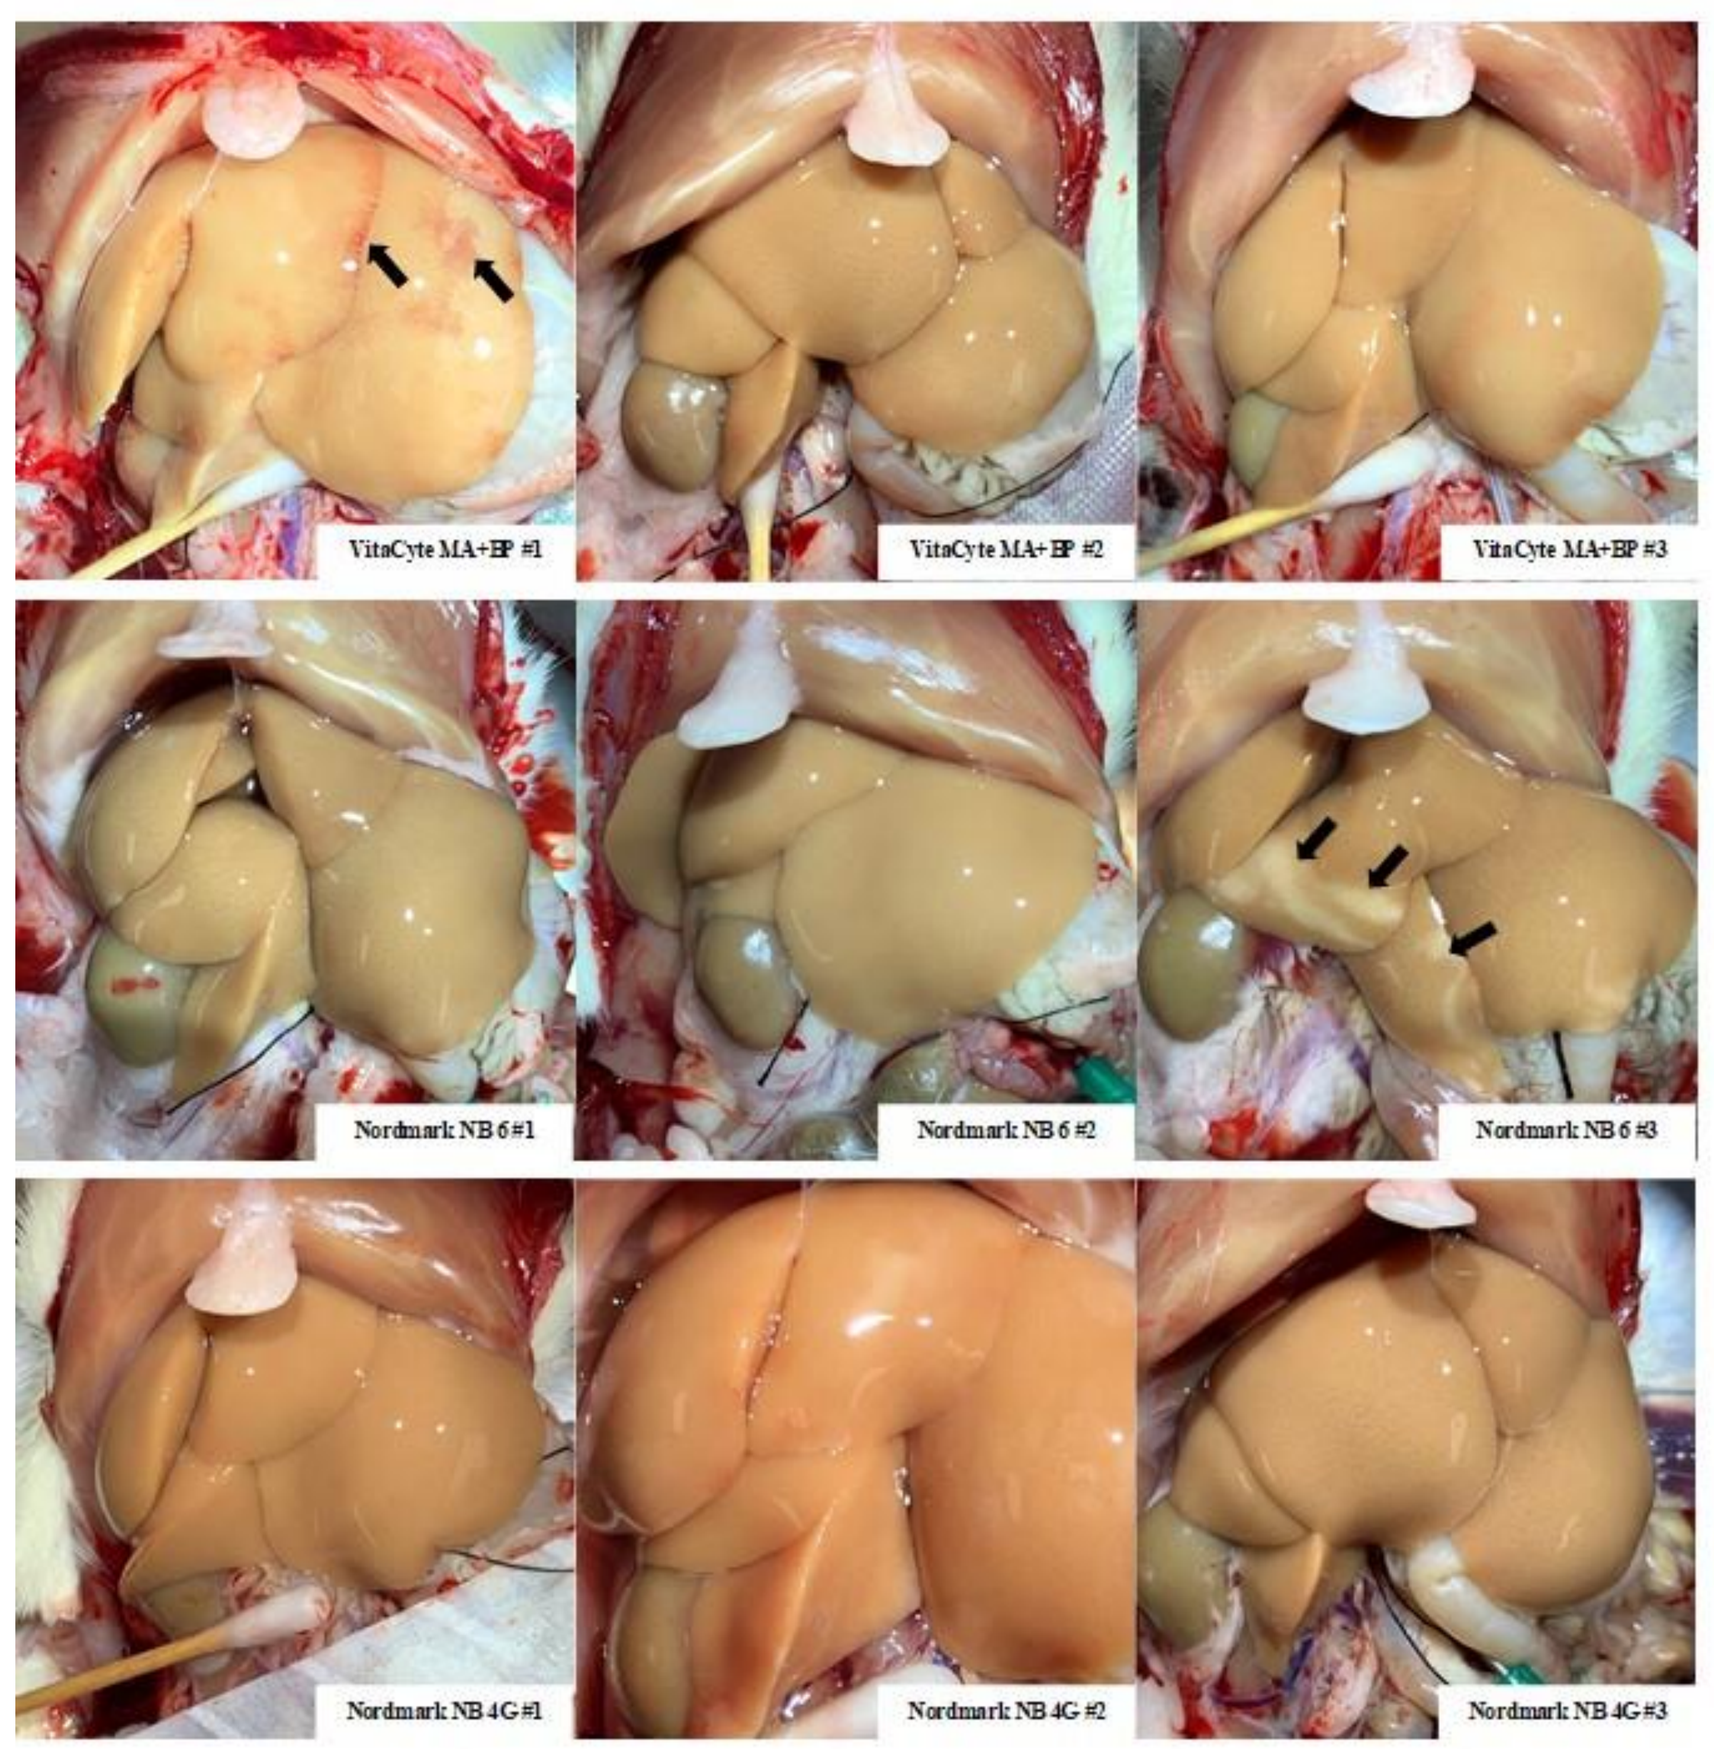
VitaCyte MA+BP #1
VitaCyte MA+BP #2
VitaCyte MA+BP #3
Nordmark NB 6 #1
Nordmark NB 6 #2
Nordmark NB 6 #3
Nordmark NB 4G #1
Nordmark NB 4G #2
Nordmark NB 4G #3

Fig 1. *In situ* perfusion. Photos were taken approximately 11 minutes after start of perfusion with the collagenase buffer solution. Each liver was perfused at 24 mL/min for approximately 15 minutes with Ca2+-free Liver Perfusion Solution I followed by perfusion with Liver Perfusion Solution II containing enzyme for 13 minutes. Arrows indicate areas of incomplete flushing of tissues.

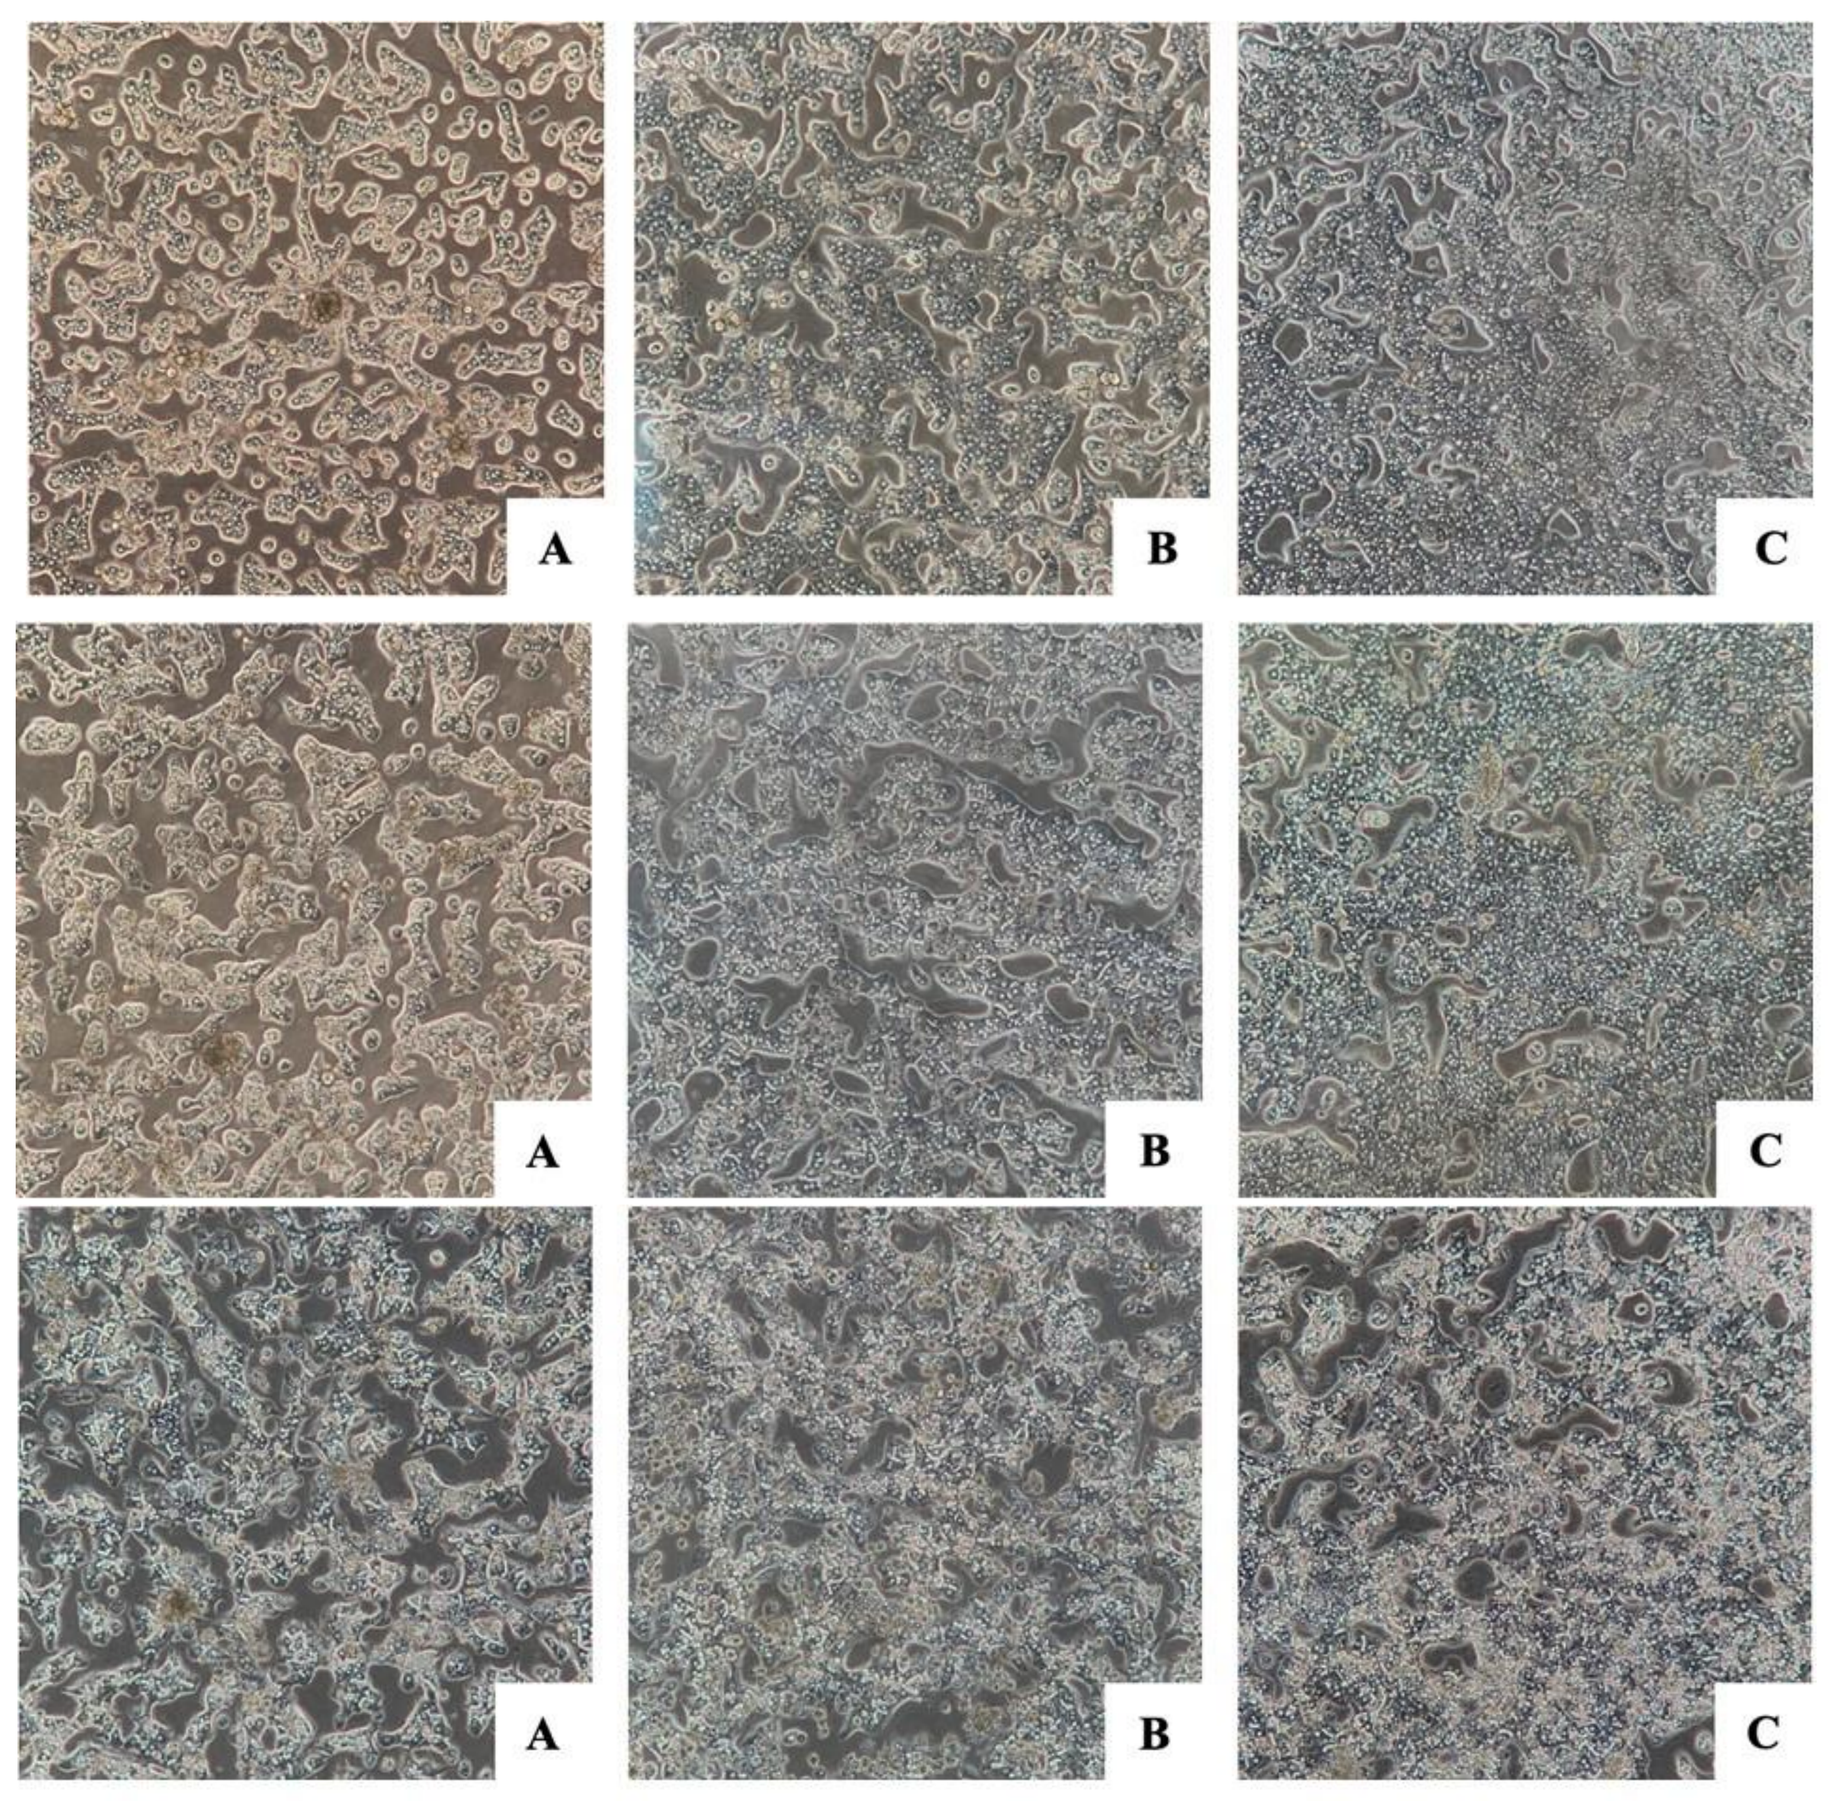
A
B
C
A
B
C
A
B
C

Fig 2. Phase-contrast images of hepatocyte cell cultures at 24 hr (Top), 48 hr (Middle), and 72 hr (Bottom). Cells isolated using **(A)**: GMP-grade VitaCyte® enzymes, **(B)**: GMP-grade Nordmark® NB 6, and **(C)**: research-grade Nordmark® NB 4G. Hepatocytes were plated at a density of $2.11x10^5$ cells/cm^2. Images were taken at 10x objective.

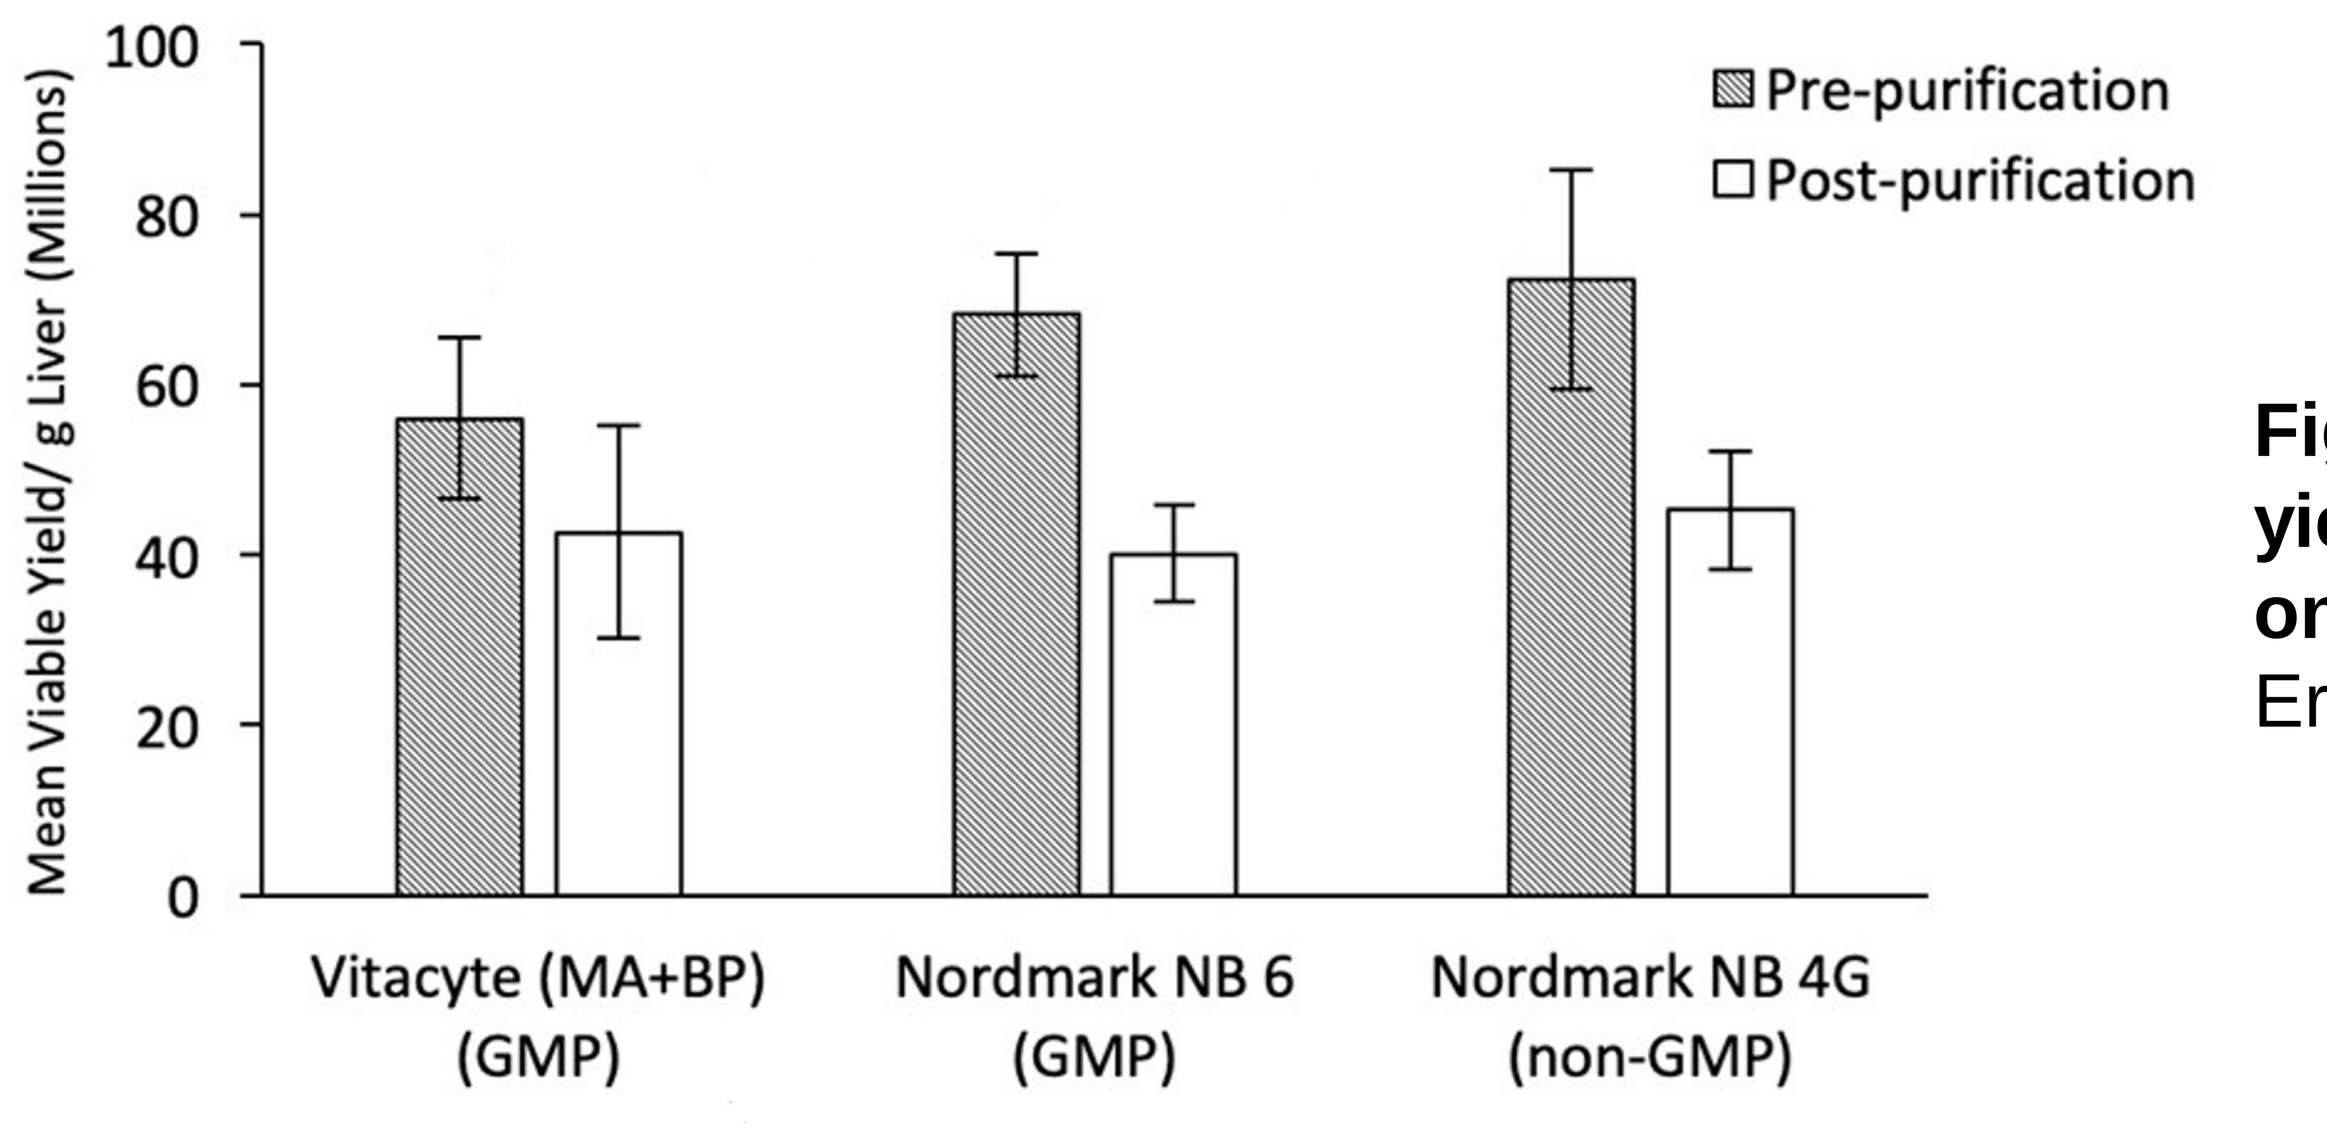
Mean Viable Yield/ g Liver (Millions)
100
80
60
40
20
0
Pre-purification
Post-purification
Vitacyte (MA+BP) (GMP)
Nordmark NB 6 (GMP)
Nordmark NB 4G (non-GMP)

Fig 3. Mean pre- and post-purification viable yields of hepatocytes per gram liver based on digestion enzyme manufacturer type. Error bars are mean ± SEM.

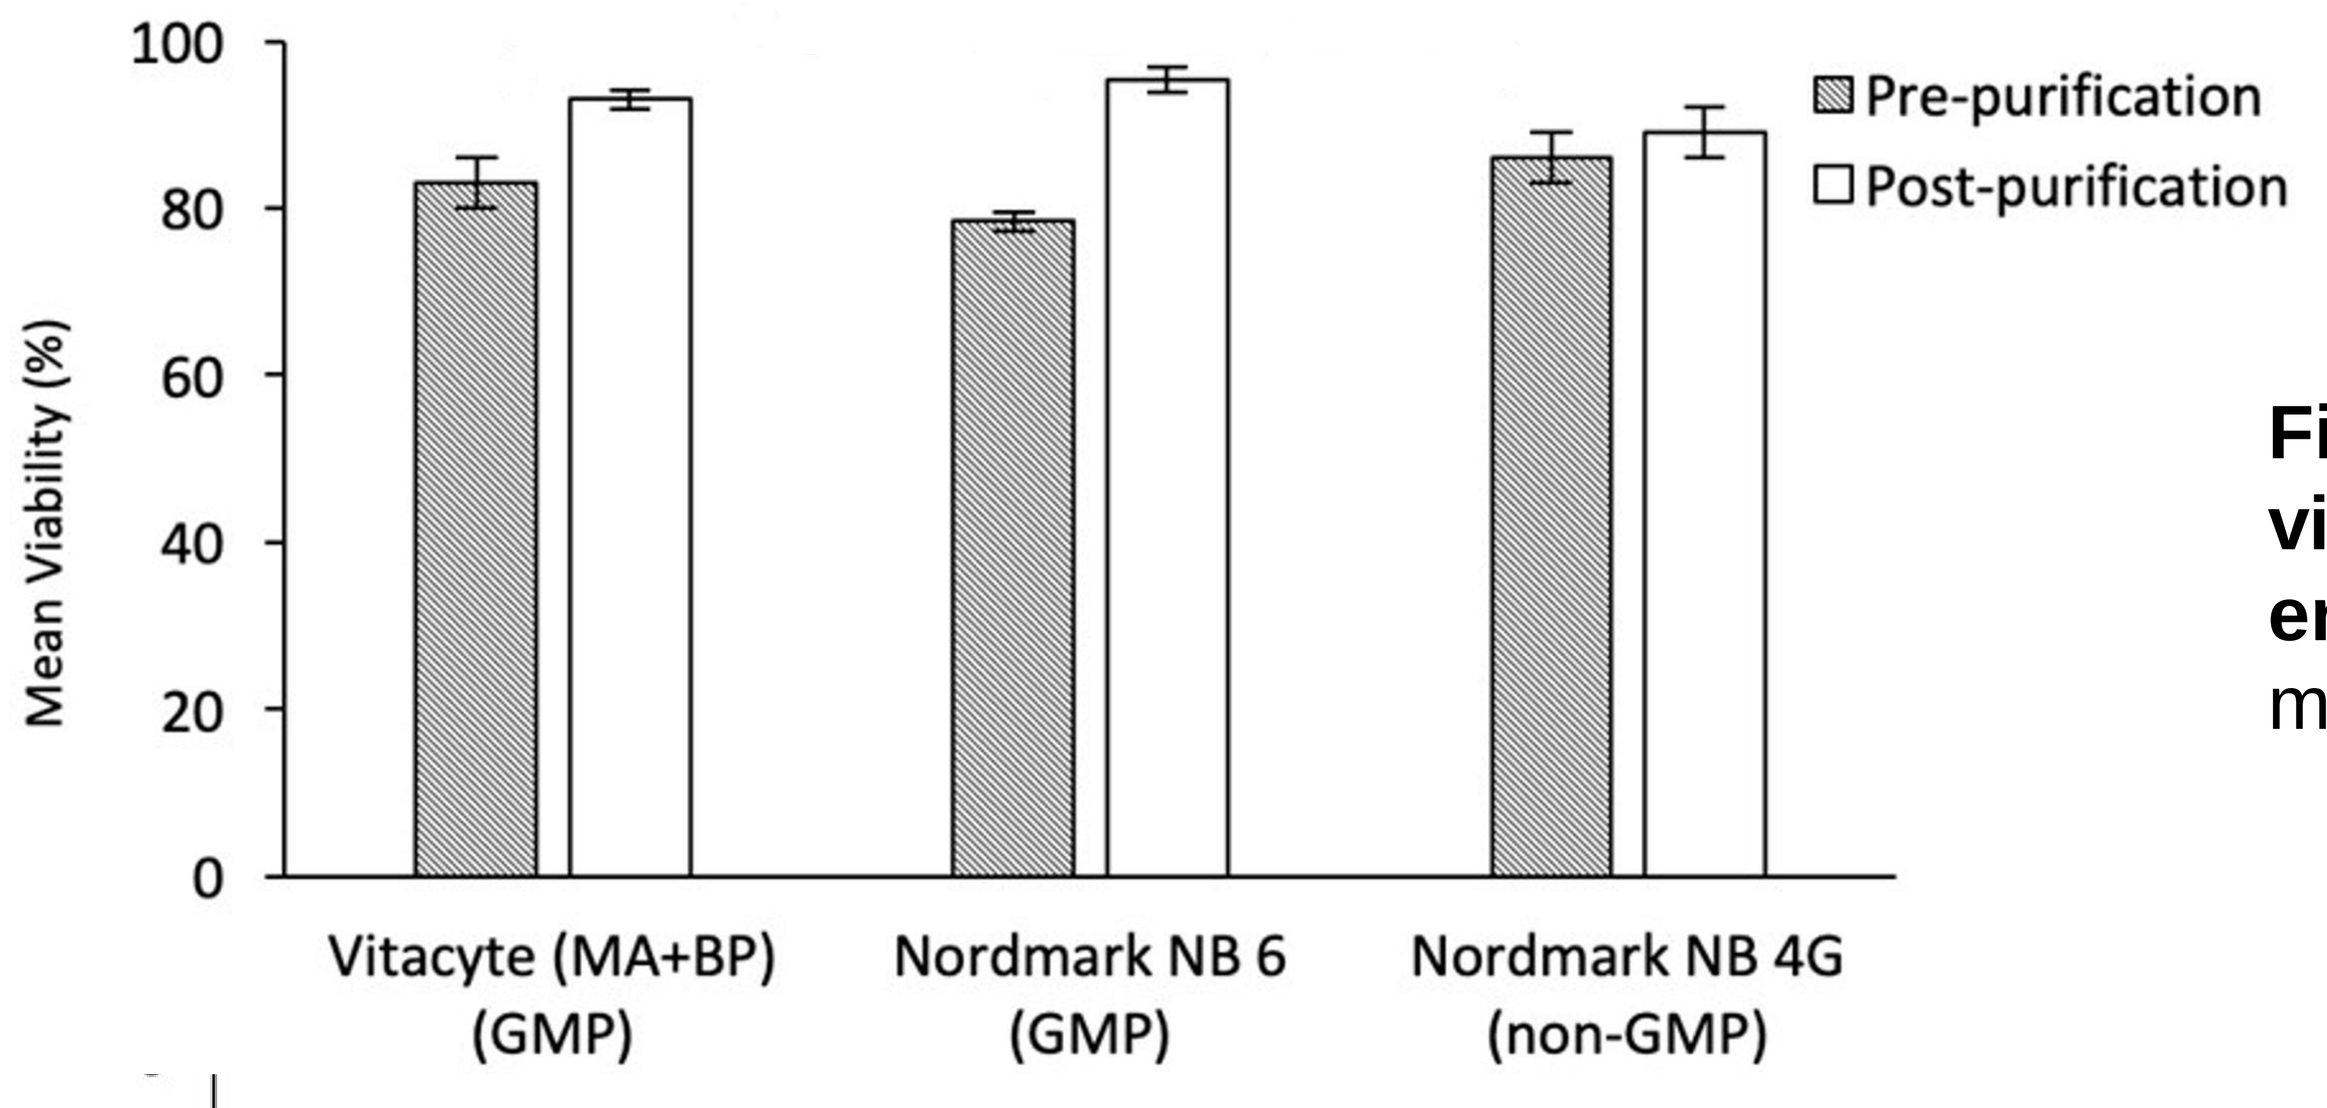
Mean Viability (%)
100
80
60
40
20
0
Pre-purification
Post-purification
Vitacyte (MA+BP) (GMP)
Nordmark NB 6 (GMP)
Nordmark NB 4G (non-GMP)

Fig 4. Mean pre- and post-purification viability of hepatocytes based on digestive enzyme manufacturer type. Error bars are mean ± SEM.

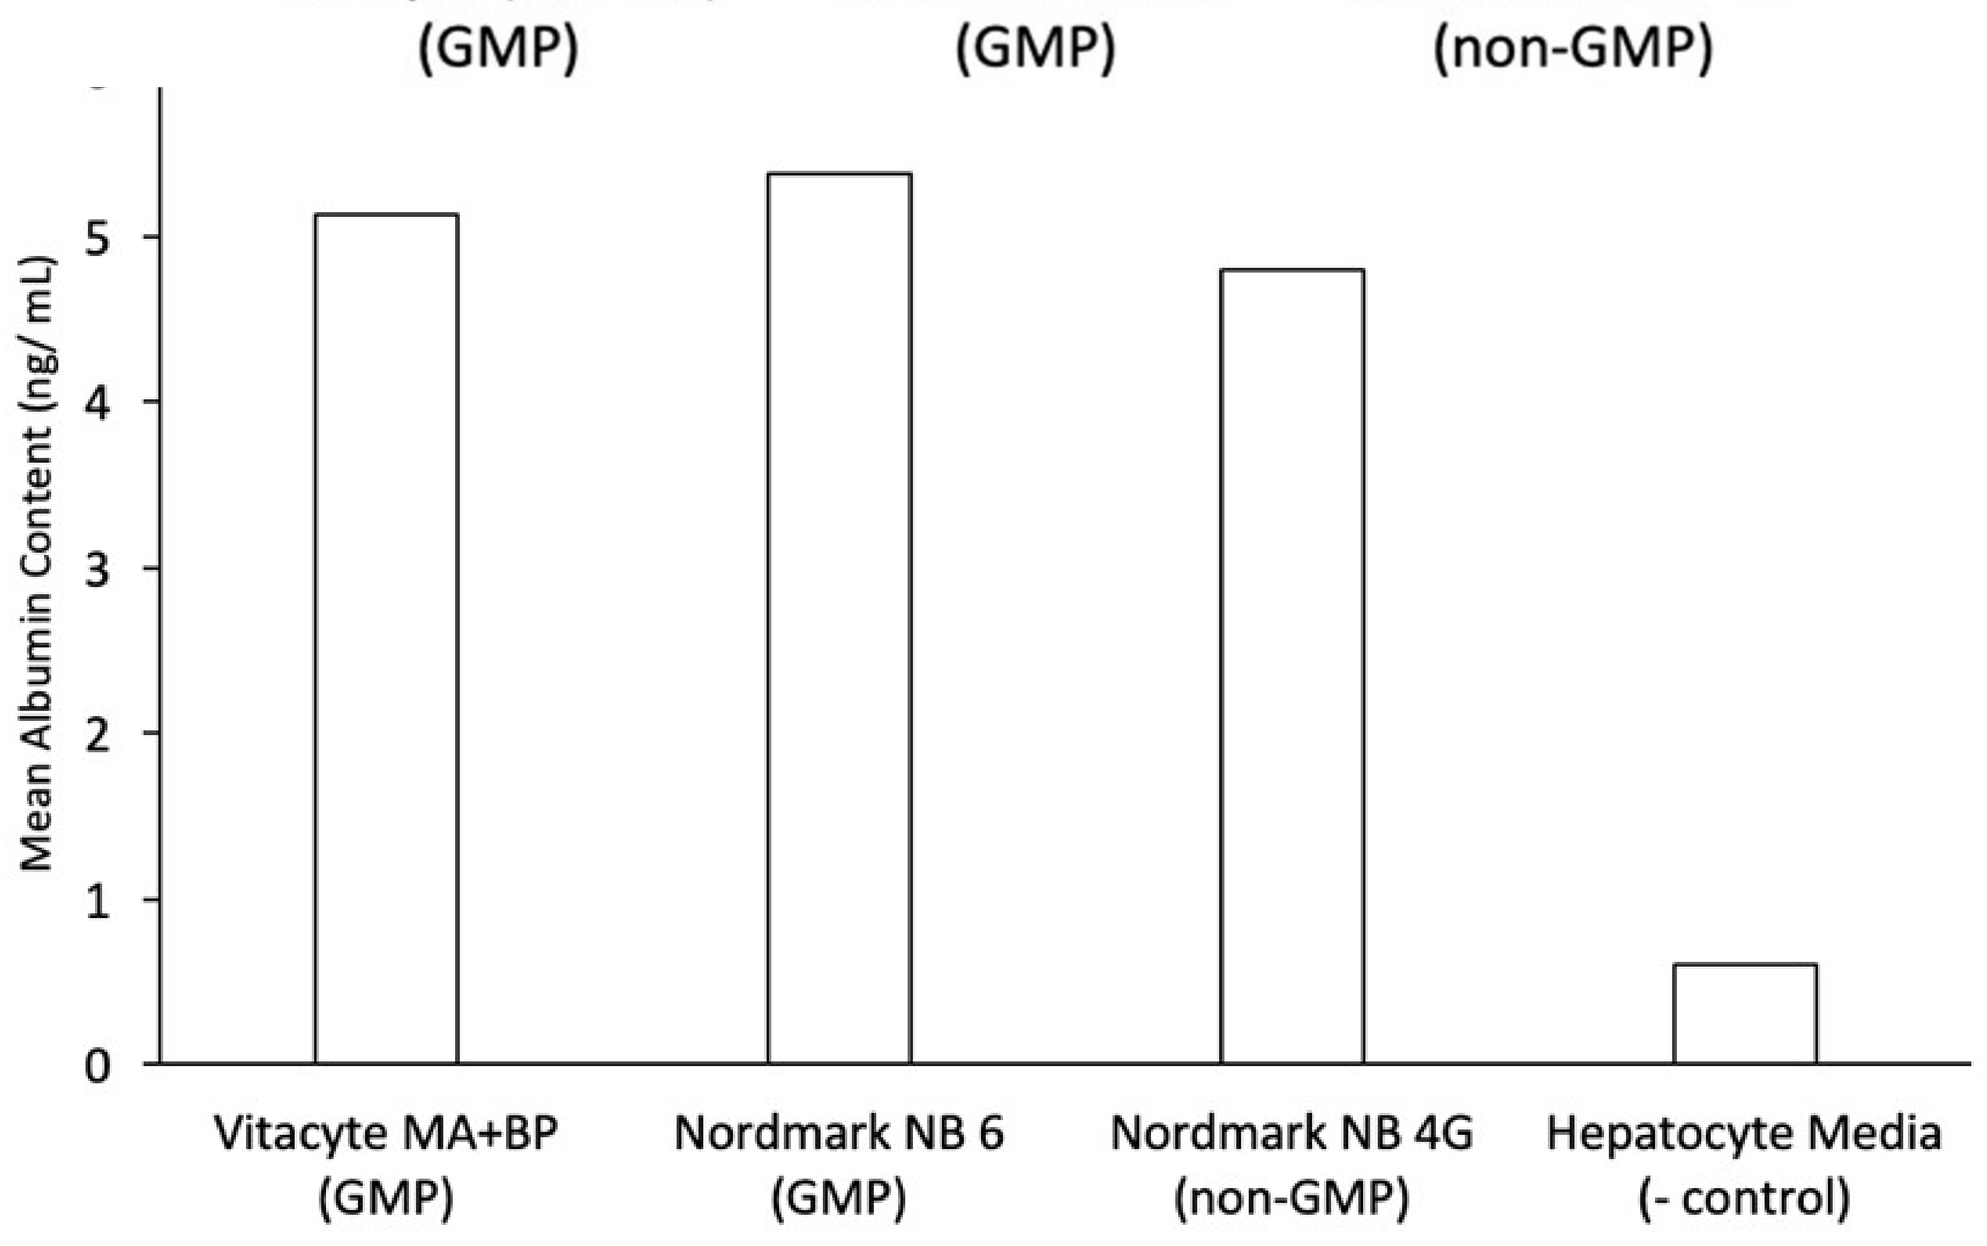
Mean Albumin Content (ng/ mL)
5
4
3
2
1
0
Vitacyte MA+BP (GMP)
Nordmark NB 6 (GMP)
Nordmark NB 4G (non-GMP)
Hepatocyte Media (- control)

Fig 5. Results from 72-hour culture media albumin ELISA testing. Data indicate the presence of albumin in culture media at 72 hours and confirm hepatocyte cell identification (n=2). Mean albumin content was consistent in media used in hepatocyte culture in the VitaCyte®, Nordmark® NB 6, Nordmark® NB 4G groups, and negative control groups, respectively (n=2).

		Pre-purification				Post-purification			
Enzyme	Mean liver mass (g)	Total yield (billion)	Viable yield (billion)	Viability (%)	Viable yield/g liver (million)	Total yield (billion)	Viable yield (billion)	Viability (%)	Viable yield/g liver (million)
VitaCyte® (MA+BP)	13.9	0.918	0.770	83.3	56.0	0.618	0.577	93.1	42.6
Nordmark® NB 6	15.4	1.21	0.951	78.4	68.2	0.650	0.619	95.4	40.1
Nordmark® NB 4G	14.7	1.20	1.05	87.0	72.4	0.745	0.657	89.0	45.3

Table 1. Pre- and post-purification yields and viability of hepatocytes by collagenase blend manufacturer. All data represent mean values (n=3).

CONCLUSION

- Nordmark® Collagenase NB 6 and VitaCyte® Collagenase MA + BP blend produced hepatocytes with comparable pre- and post-purification viable yields and viability.
- Non-GMP-grade Nordmark® Collagenase NB 4G furnished primary hepatocytes with pre- and post-purification yields and viability not significantly different from GMP-grade Nordmark® Collagenase NB 6.
- GMP-grade Collagenase Nordmark® NB 6 is a viable alternative to GMP-grade VitaCyte® Collagenase MA+BP enzyme blend for primary hepatocyte isolation, and non-GMP-grade Nordmark® Collagenase NB 4G is an affordable alternative for hepatocyte application in research.
- GMP-grade VitaCyte® and Nordmark® enzymes are manufactured to cGMP standards and can therefore be utilized in the development of protocols intended for clinical application.

ACKNOWLEDGMENTS

This study was conducted with material support from Nordmark Biochemicals and funding from Ambys Medicines (grant #AM-5572993), as well as support from UC Irvine Department of Surgery.

PUBLICATION

Japanese Journal of Gastroenterology and Hepatology. 2022, V8(20): 1-7.

www.amsbio.com