Proof of Concept for Discovering Allosteric Kinase Inhibitors by Characterising a MEK1 Type III Kinase Inhibitor Using a Fluorescent ATP Analogue

Oliver Medhurst, Sabine Milhas, Su Jerwood, Martin Redhead; *Exscientia*

INTRODUCTION

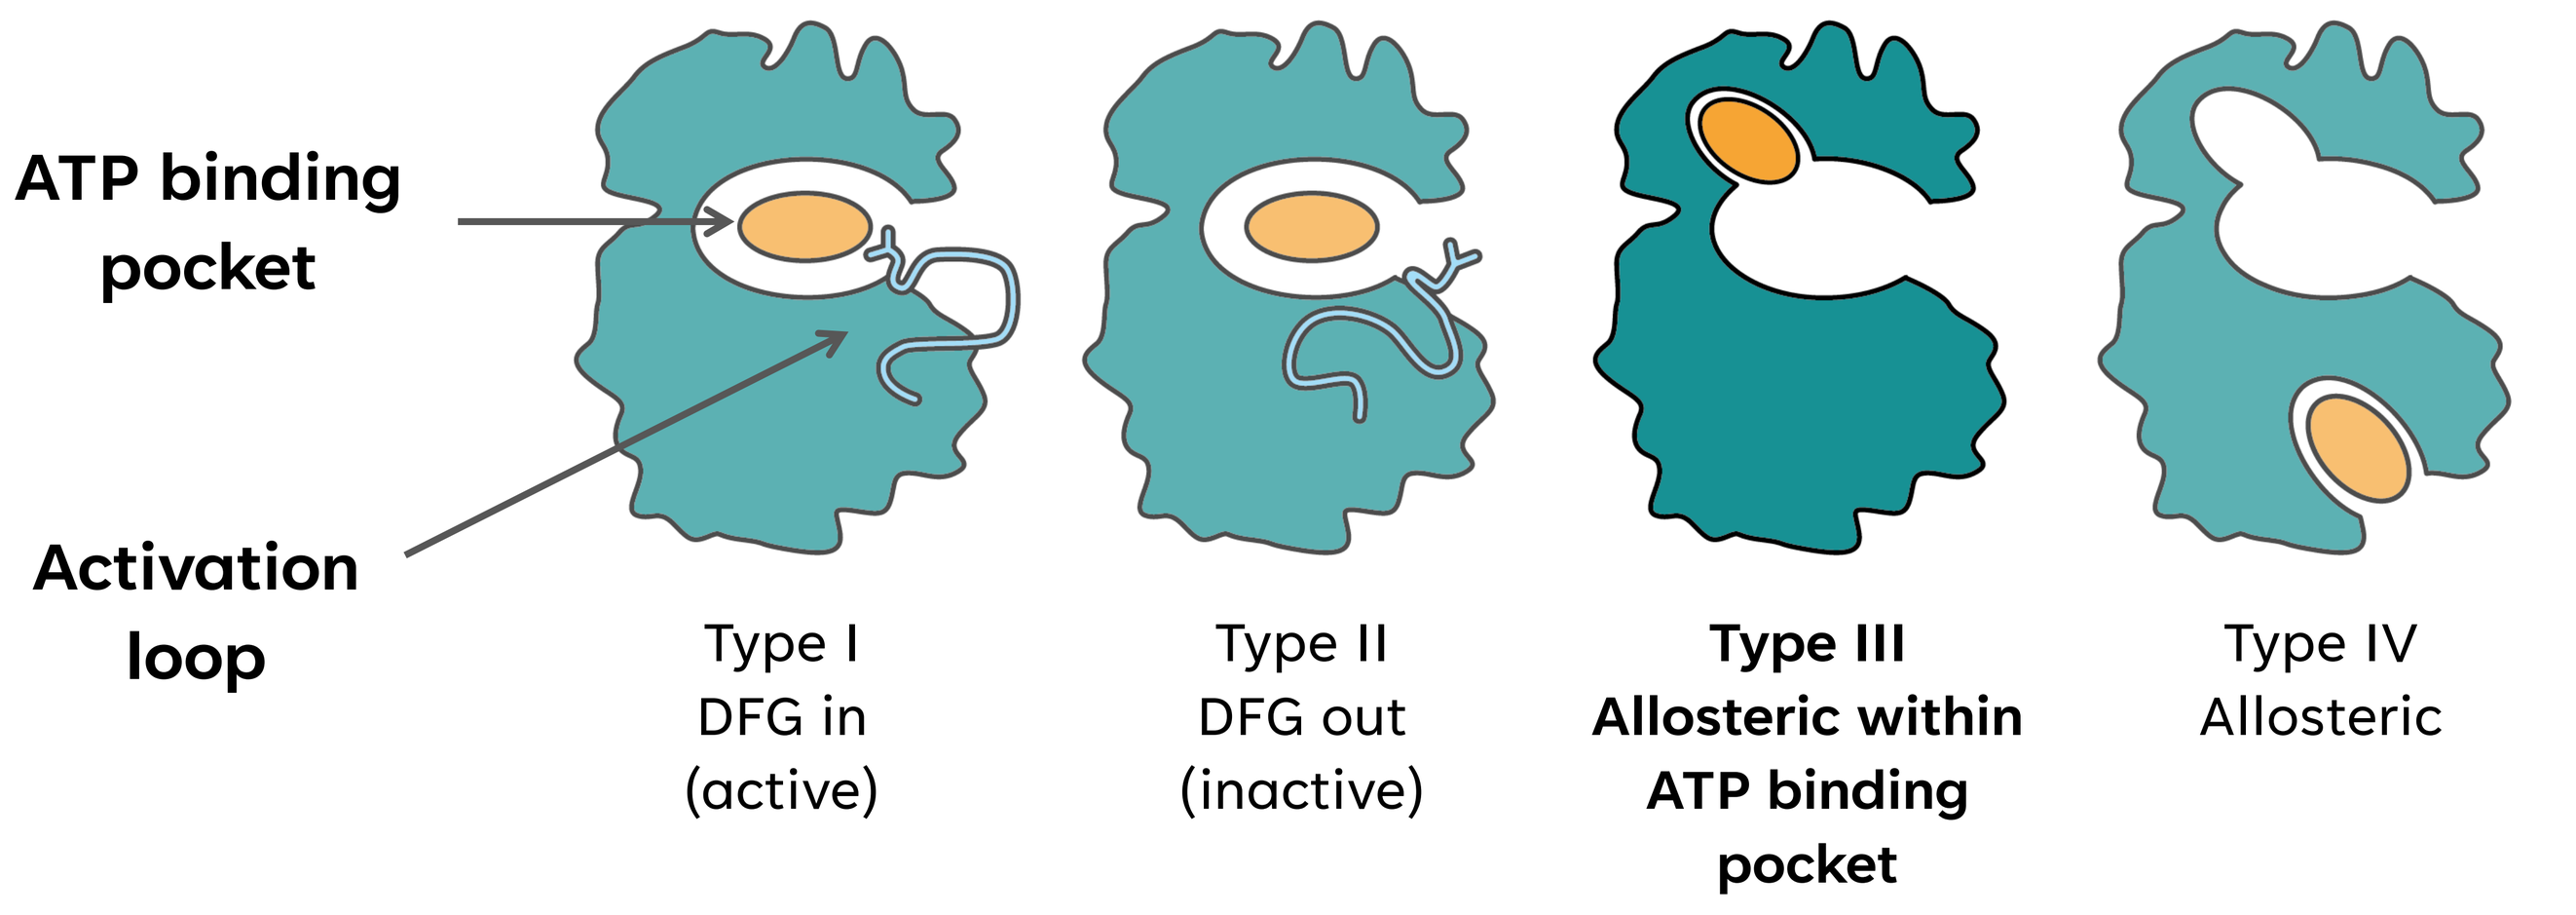
ATP binding pocket
Activation loop
Type I
DFG in
(active)
Type II
DFG out
(inactive)
Type III
Allosteric within ATP binding pocket
Type IV
Allosteric

Small-molecule kinase inhibitors are an important cancer therapy approach, many targeting the highly conserved ATP binding pocket to block ATP and kinase interactions. However, low selectivity and gatekeeper mutation development ultimately leads to acquired resistance, driving a new search for allosteric kinase inhibitors. Type III kinase inhibitors bind to target kinases allosterically within the catalytic domain, adjacent to the ATP binding-site. Once bound, these inhibitors allow and promote ATP binding, which we have leveraged to develop an assay to detect type III inhibitors using a fluorescent ATP analogue.

METHODS

Assay: GST-tagged MEK1 protein prepared in magnesium rich assay buffer was added to 384-well assay plates containing test inhibitor PD0325901 (mirdametinib), and pre-incubated for 30 min at room temperature (RT). Fluorescent non-hydrolysable ATP analogue was added, followed by a 30 min RT incubation. Finally, terbium-labelled anti-GST antibody was applied to the assay plate followed by a 2 hour RT incubation. The assay plate was read using the PHERAstar FSX plate reader on HTRF mode.

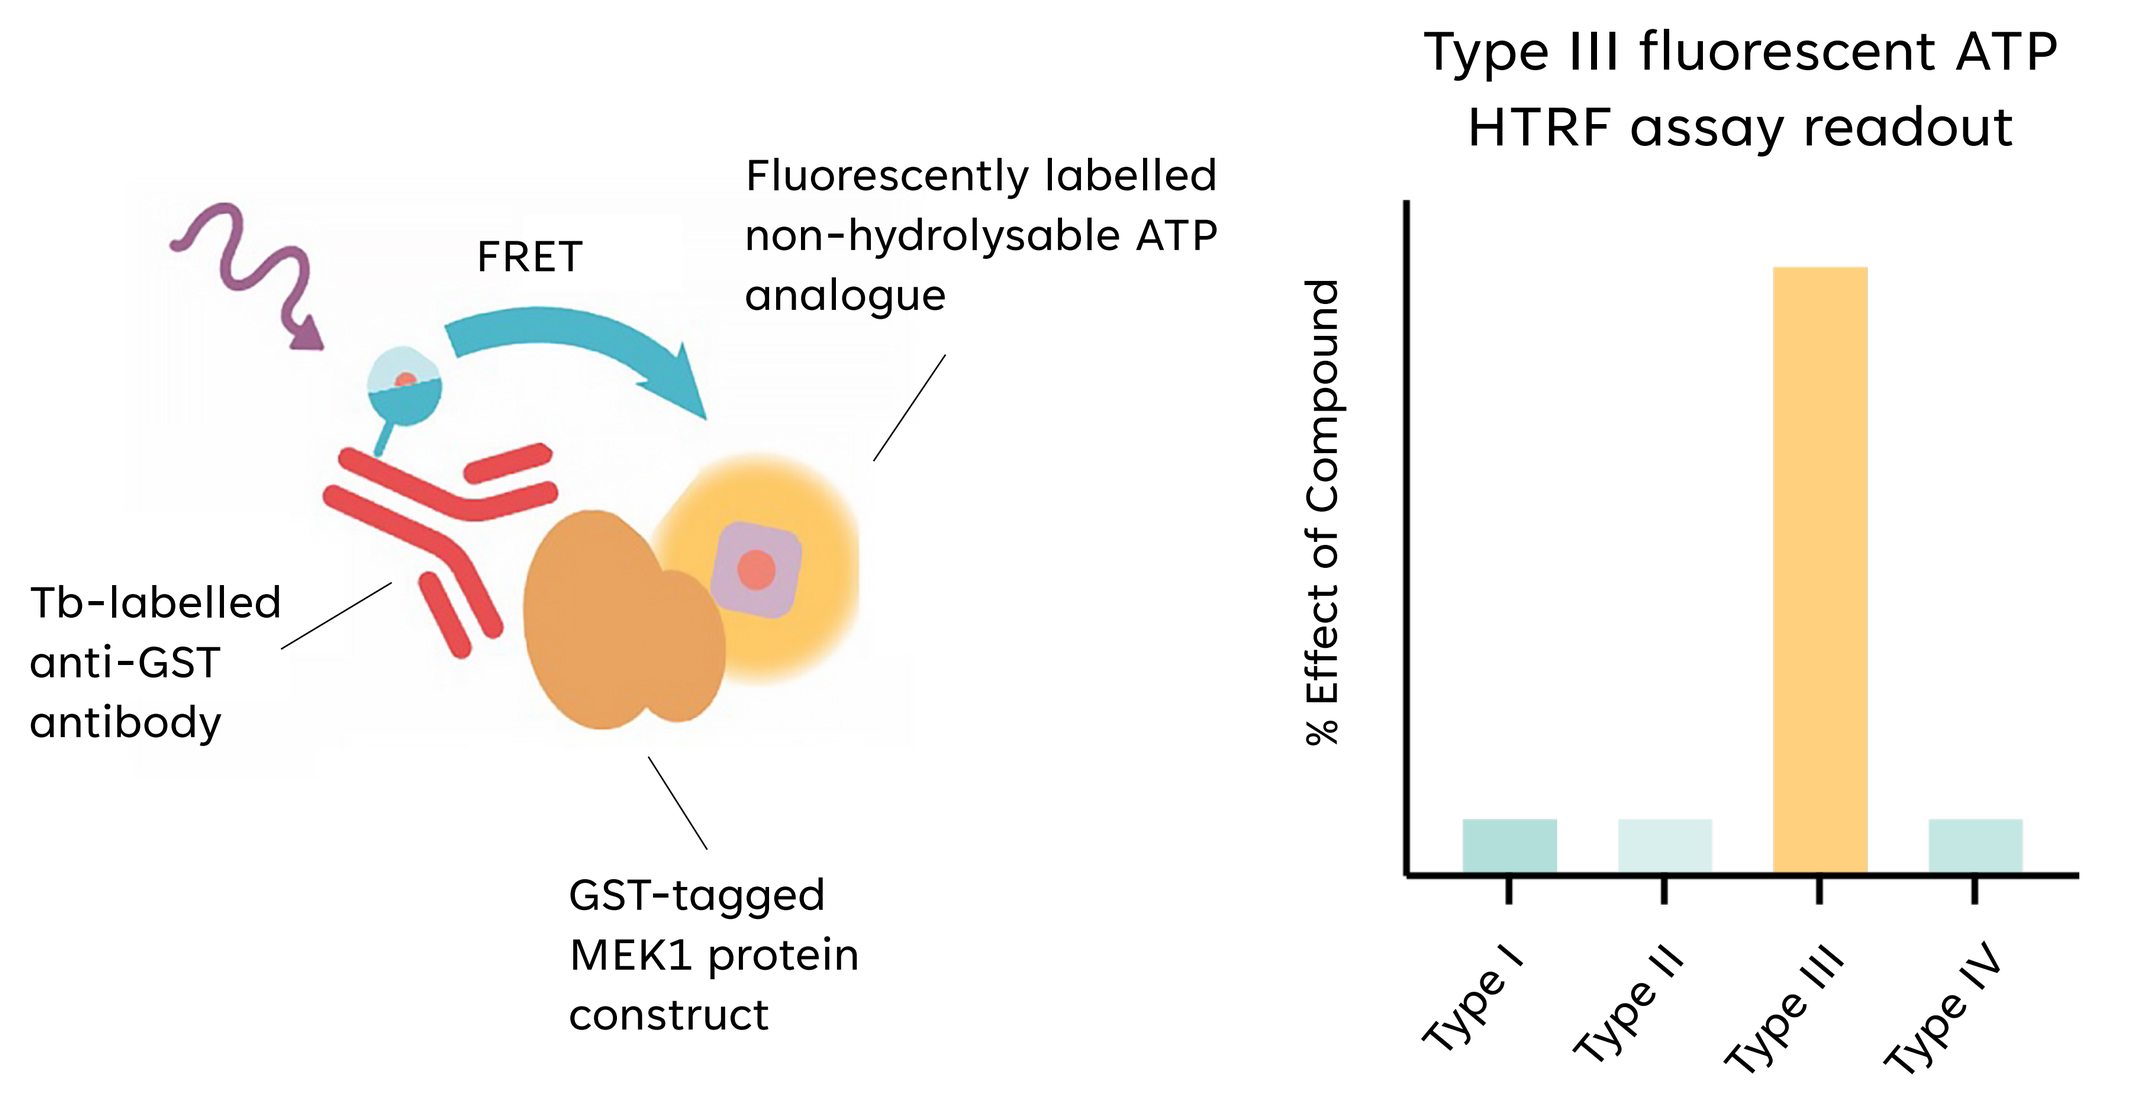
FRET
Fluorescently labelled non-hydrolysable ATP analogue
Tb-labelled anti-GST antibody
GST-tagged MEK1 protein construct
Type III fluorescent ATP HTRF assay readout
% Effect of Compound
Type I
Type II
Type III
Type IV

MEK1 HTRF binding assay schematic using non-hydrolysable fluorescent ATP analogue, which is advantageous for differentiating type III inhibitors. Schematics adapted from Cisbio.

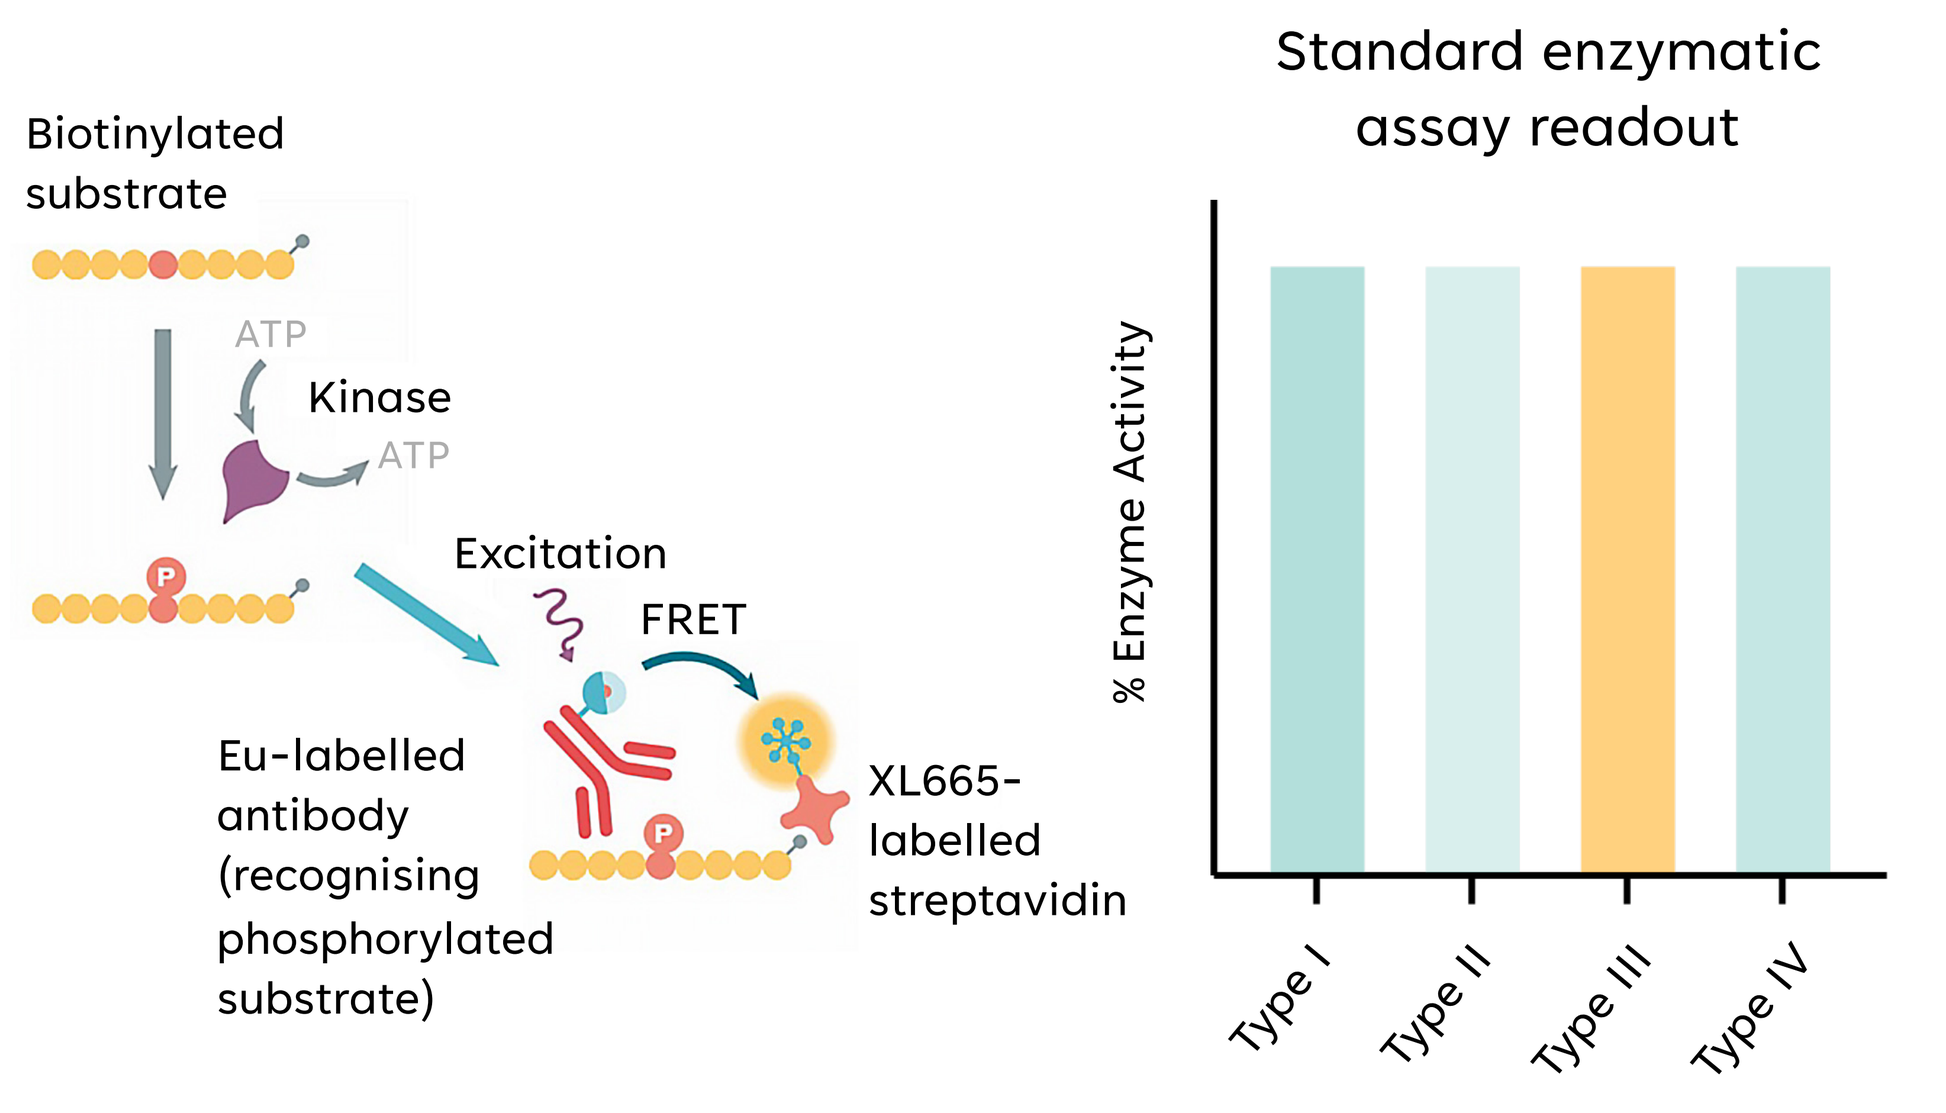
Biotinylated substrate
ATP
Kinase
ATP
Excitation
FRET
Eu-labelled antibody (recognising phosphorylated substrate)
XL665-labelled streptavidin
Standard enzymatic assay readout
% Enzyme Activity
Type I
Type II
Type III
Type IV

Standard enzymatic kinase assay schematic with the limitation of similar assay readout for each kinase inhibitor type, prompting the development of more selective assays.

Data Analysis: GraphPad Prism version 9.4.1 was used to fit all dose-response curves to a four-parameter fit function and perform all statistical analysis.

RESULTS

Figure 1: Buffer optimisation enhances HTRF assay signal over 10-fold when PD0325901 is applied

A)

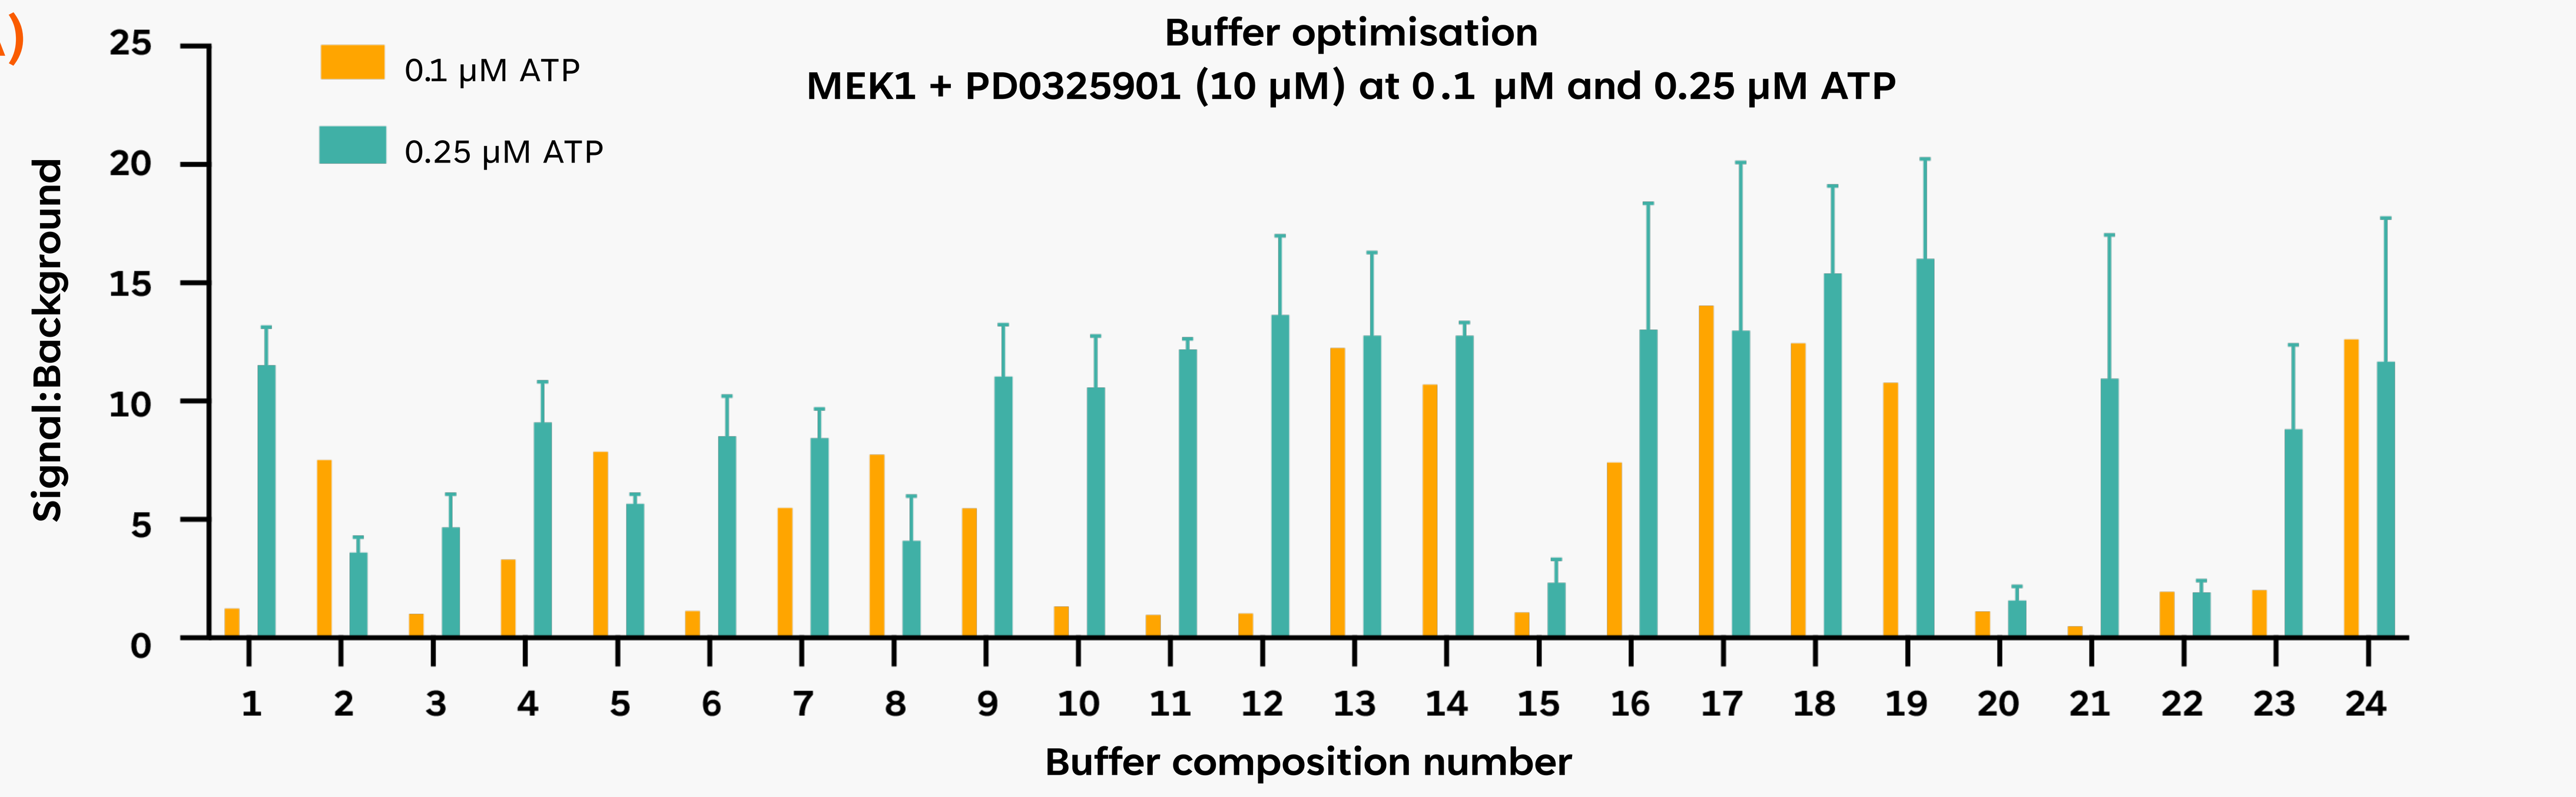
Buffer optimisation
MEK1 + PD0325901 (10 μM) at 0.1 μM and 0.25 μM ATP
0.1 μM ATP
0.25 μM ATP
Signal:Background
25
20
15
10
5
0
1 2 3 4 5 6 7 8 9 10 11 12 13 14 15 16 17 18 19 20 21 22 23 24
Buffer composition number

B)

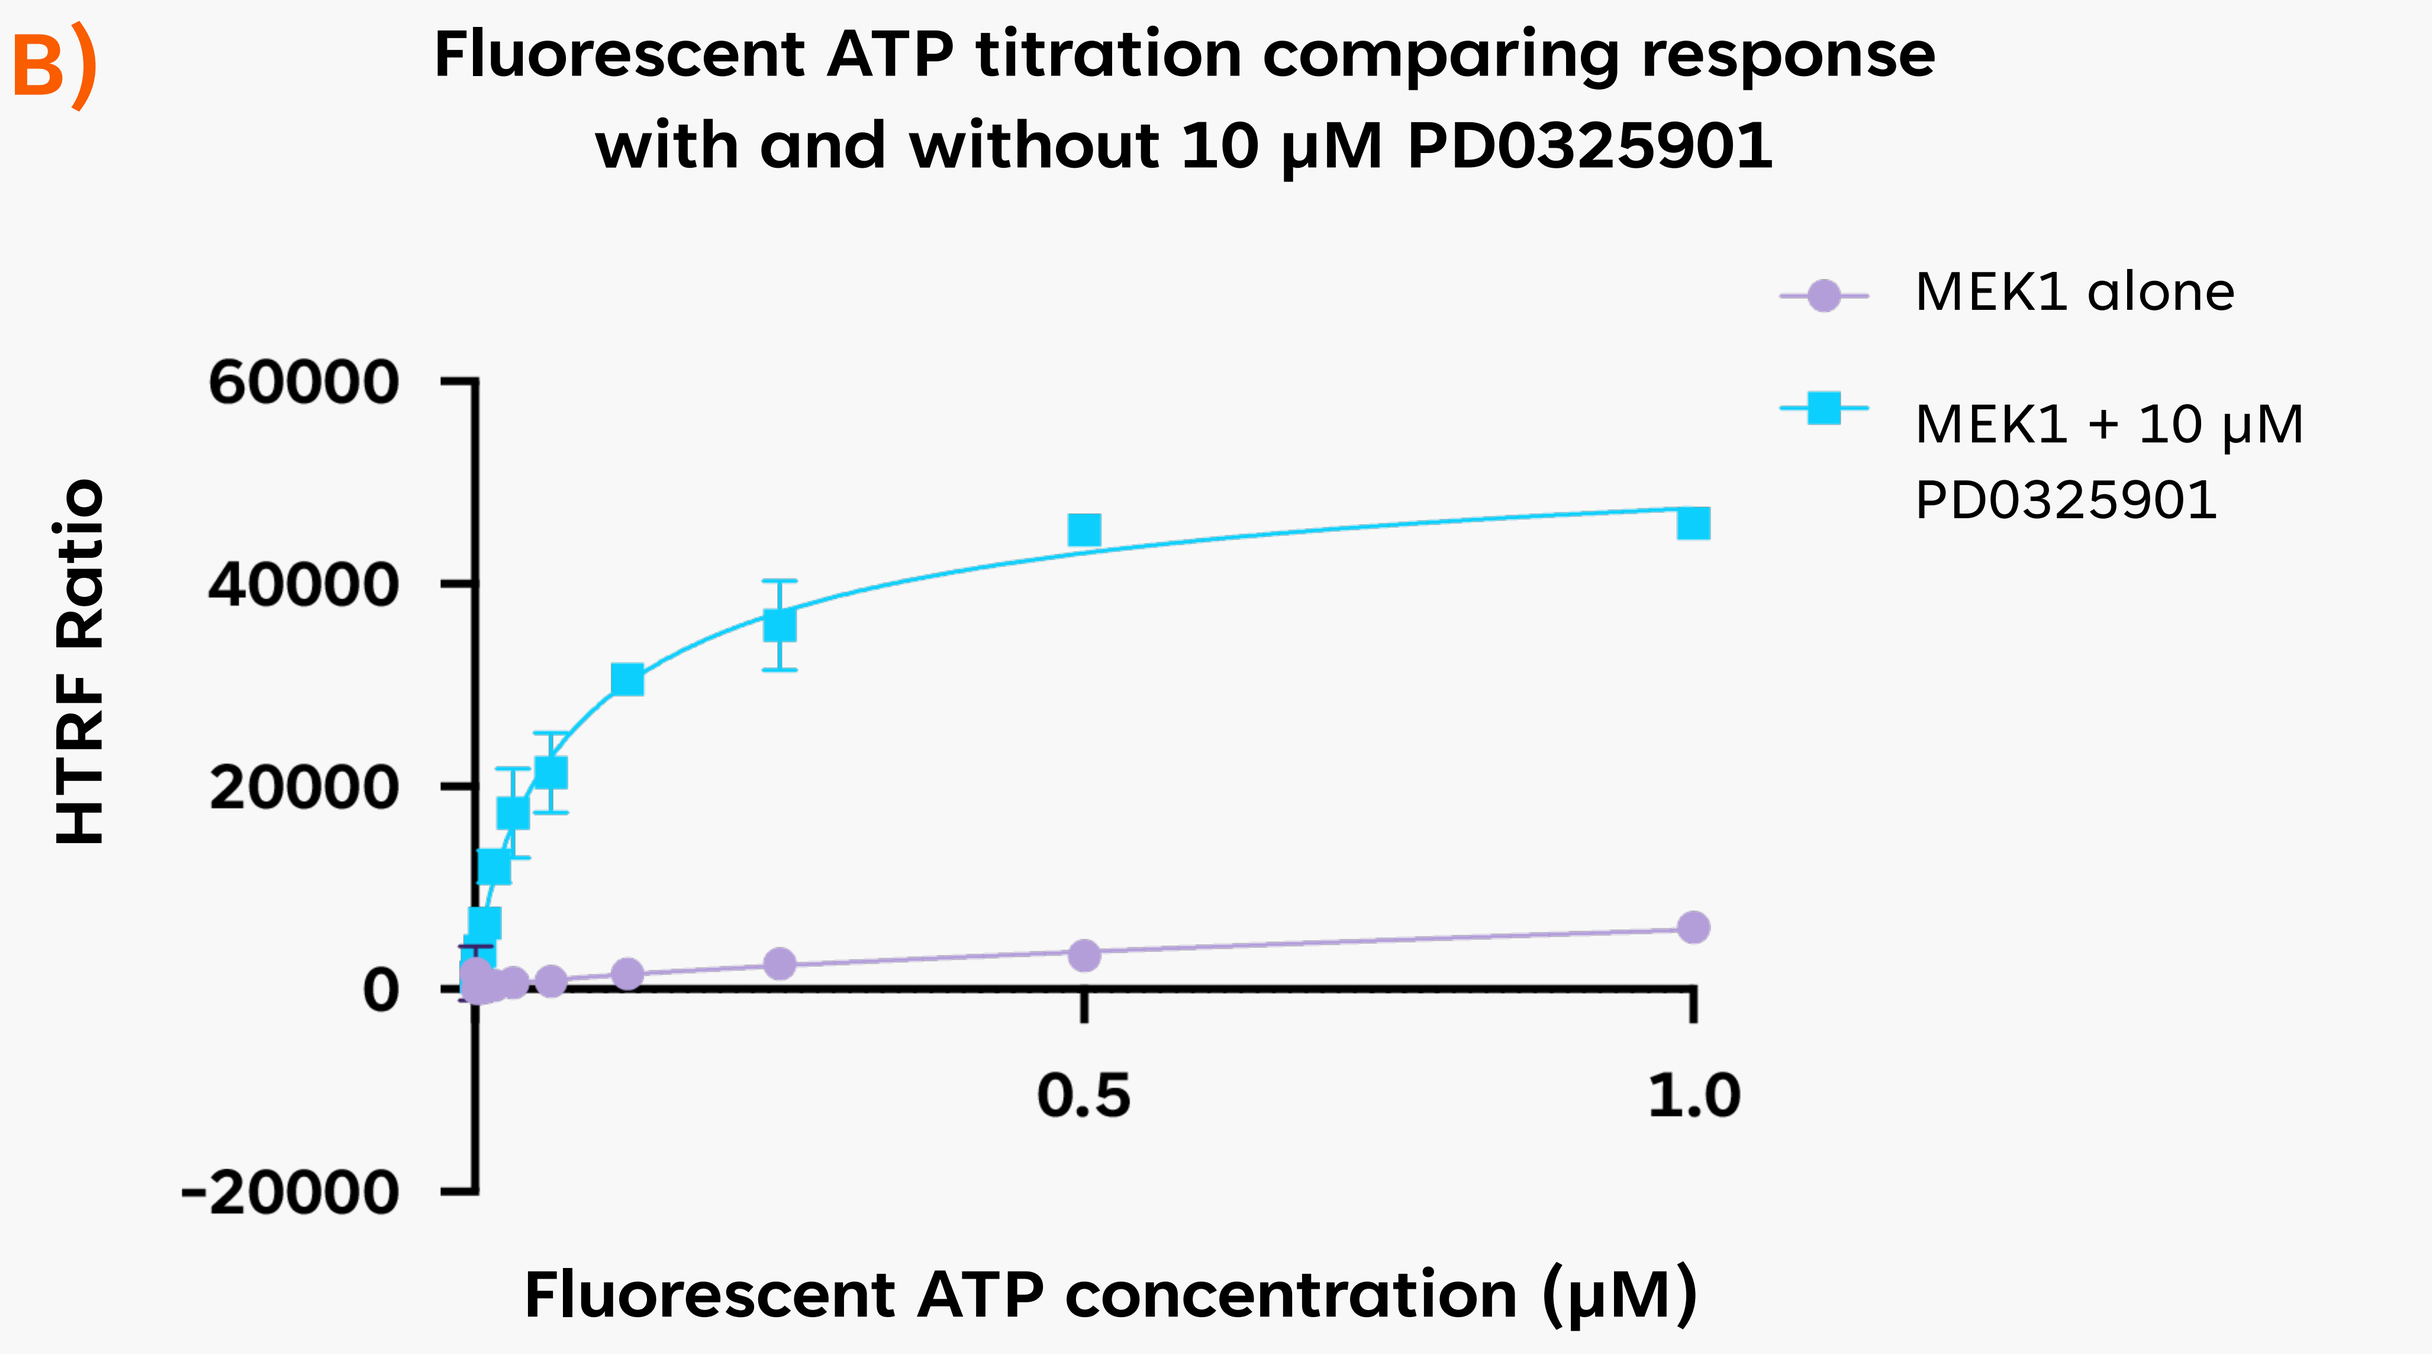
Fluorescent ATP titration comparing response with and without 10 μM PD0325901
MEK1 alone
MEK1 + 10 μM PD0325901
HTRF Ratio
60000
40000
20000
0
-20000
0.5
1.0
Fluorescent ATP concentration (μM)

Figure 1: (A) Buffer conditions were optimised to provide the highest inhibitor-dependent increase in assay signal, testing 24 different buffer compositions with various concentrations of buffer components (TCEP, Triton X-100, $MgCl_2$, NaCl & BSA) and buffering agents (phosphate, MOPS, Tris & HEPES). **(B)** In optimised buffer conditions, PD0325901 appeared to cause more than 10-fold increase in HTRF ratio when compared to MEK1 alone.

Figure 2: Dose-ratio titrations of fluorescent ATP and PD0325901 show pharmacological dependency impacting K_D and EC_{50} respectively

A1)

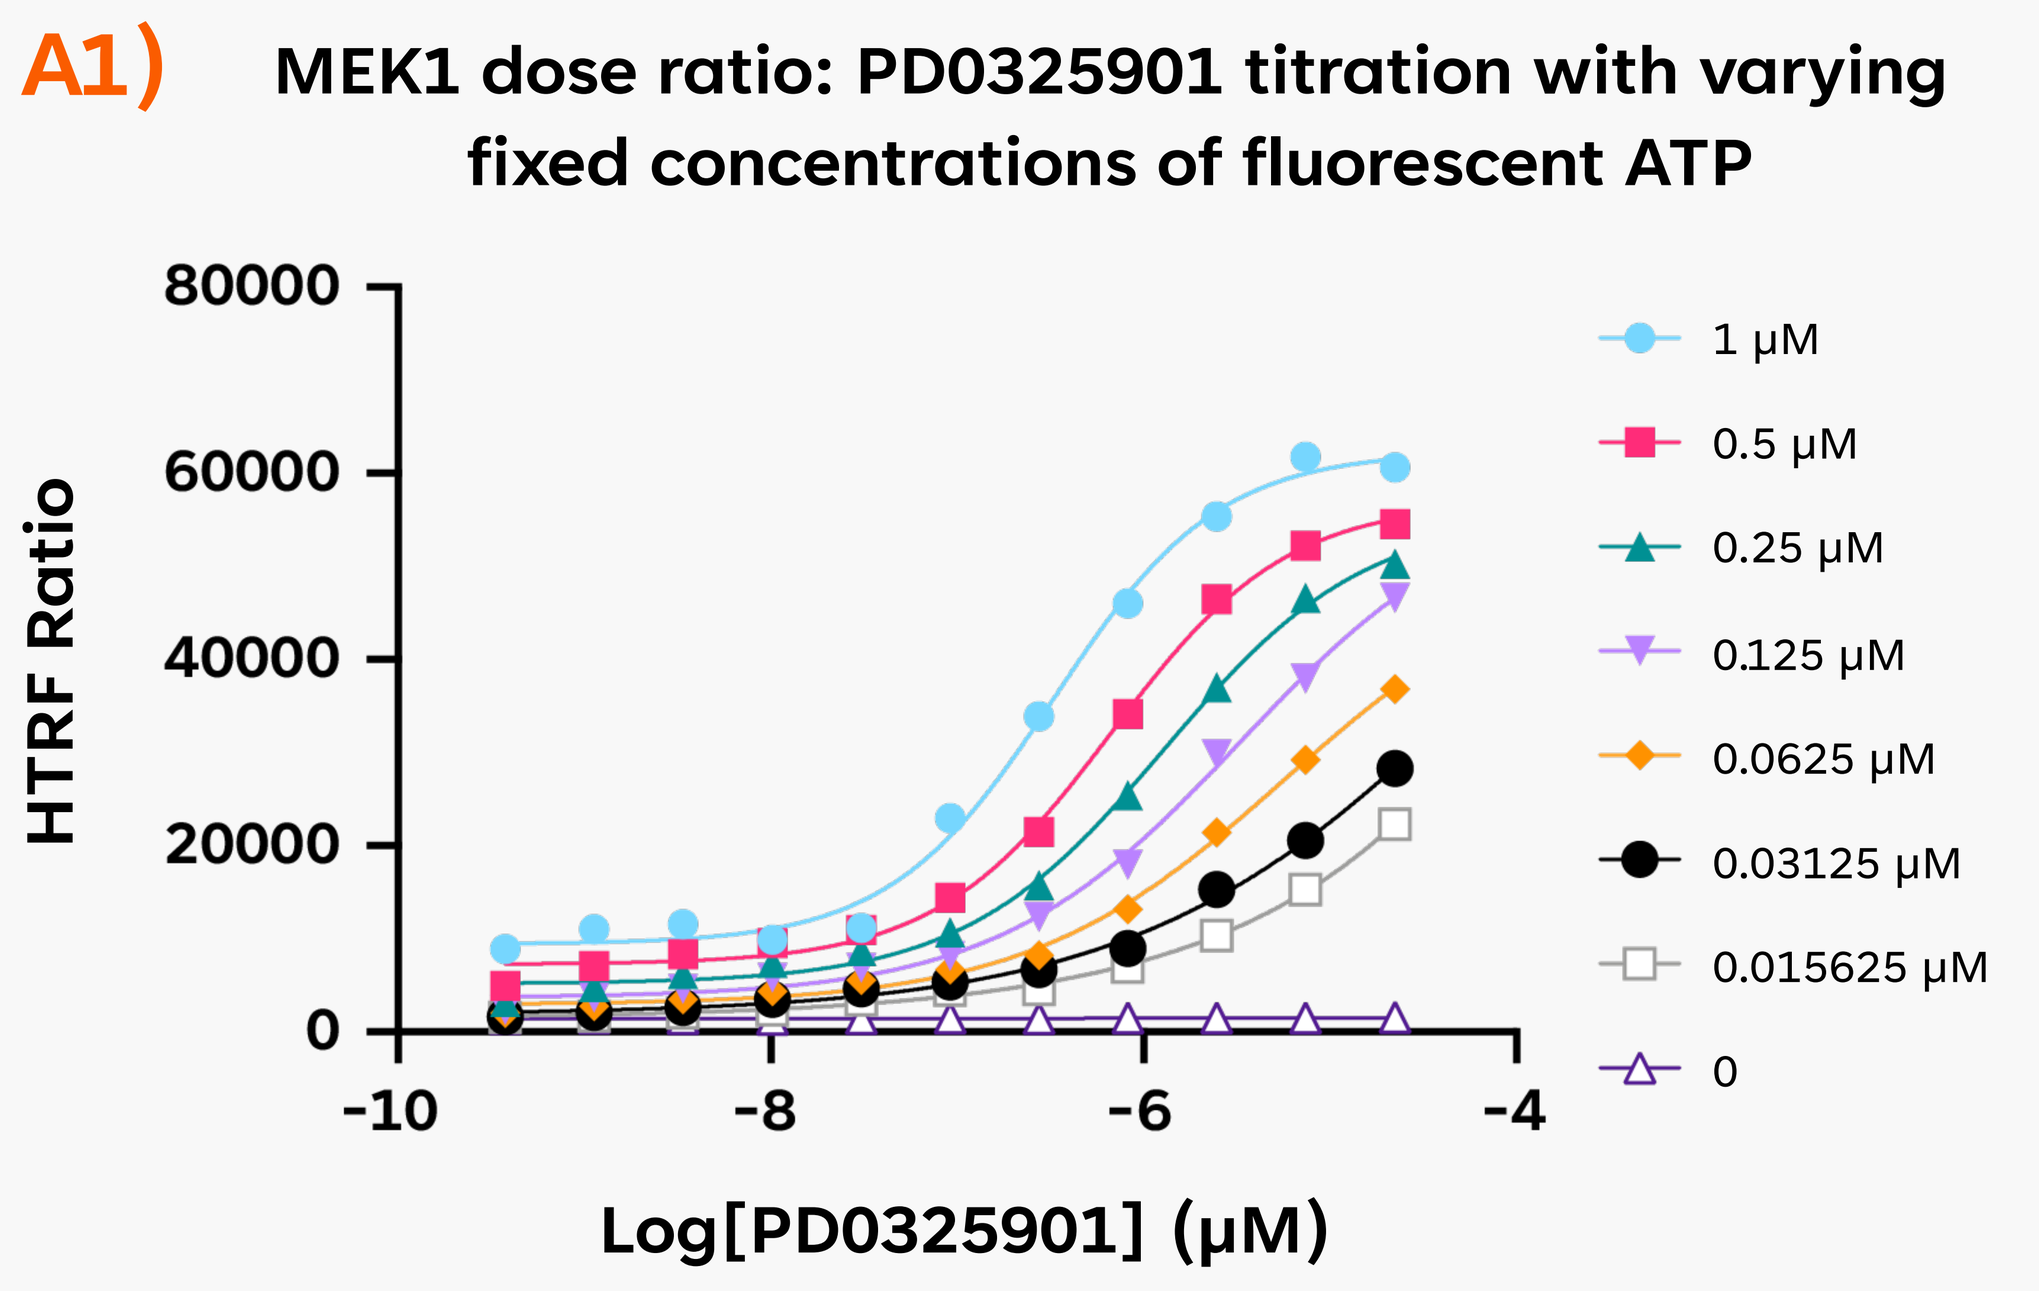
MEK1 dose ratio: PD0325901 titration with varying fixed concentrations of fluorescent ATP
HTRF Ratio
80000
60000
40000
20000
0
-10
-8
-6
-4
Log[PD0325901] (μM)
1 μM
0.5 μM
0.25 μM
0.125 μM
0.0625 μM
0.03125 μM
0.015625 μM
0

A2)

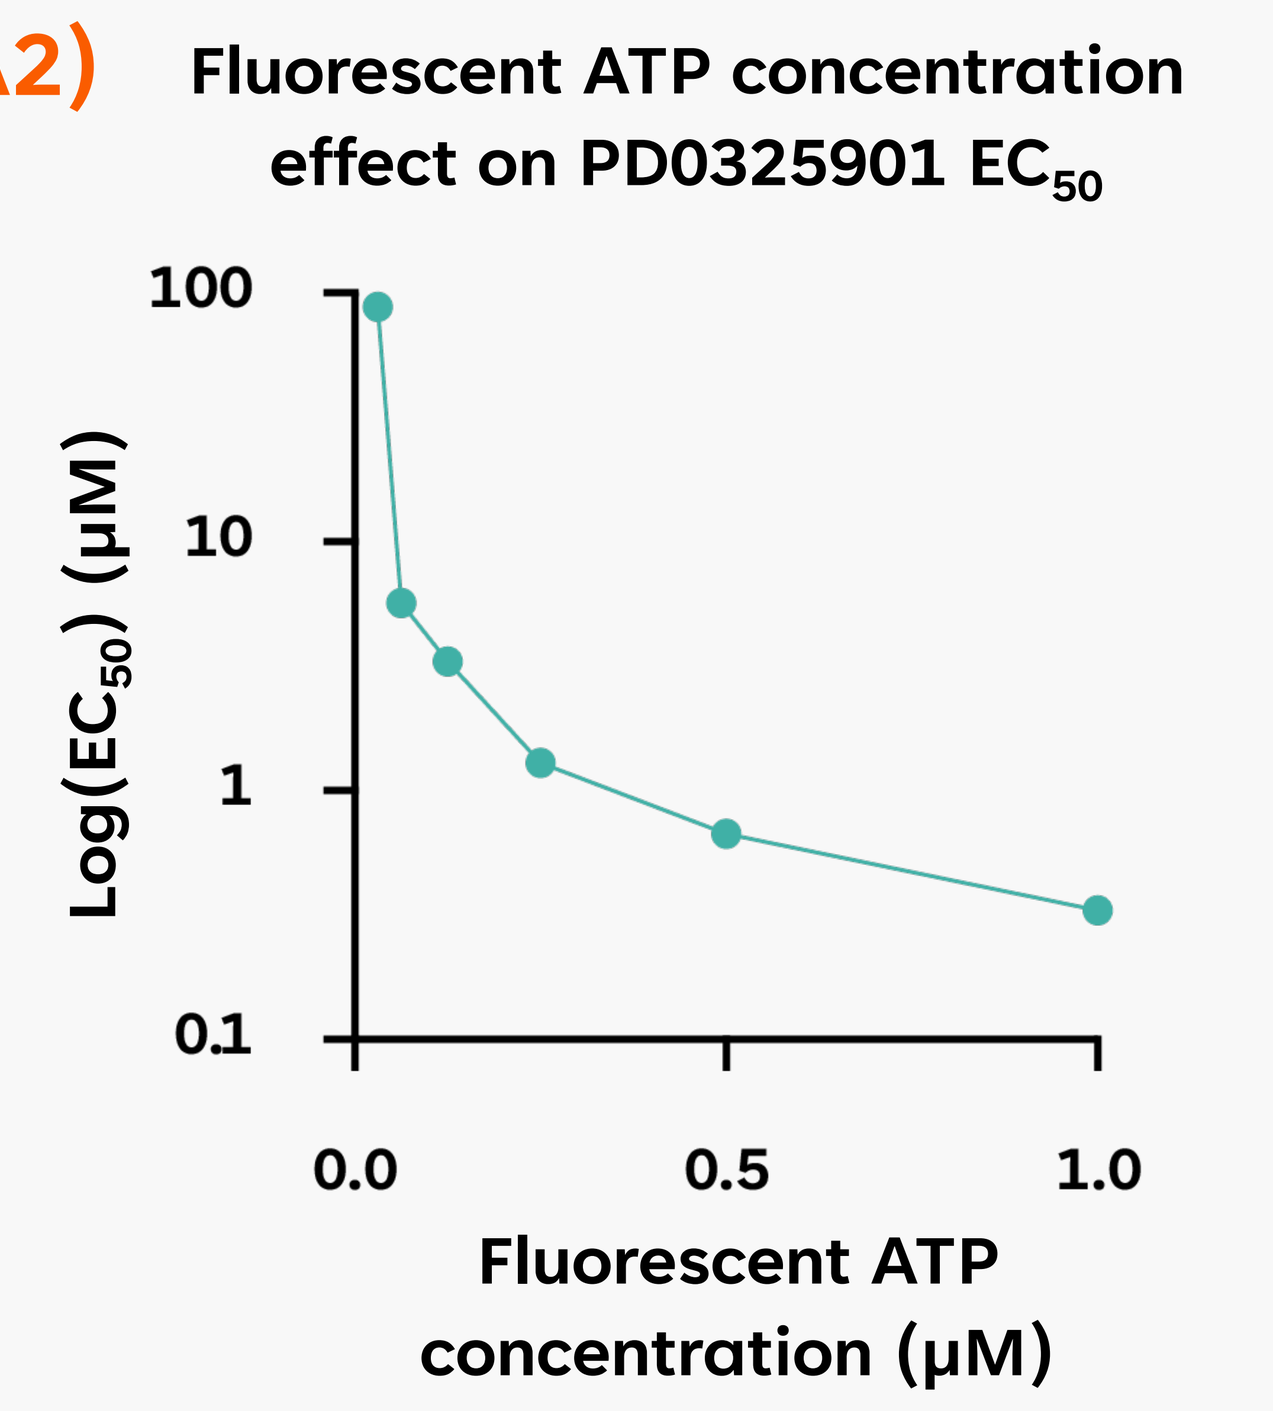
Fluorescent ATP concentration effect on PD0325901 EC50
Log(EC50) (μM)
100
10
1
0.1
0.0
0.5
1.0
Fluorescent ATP concentration (μM)

B1)

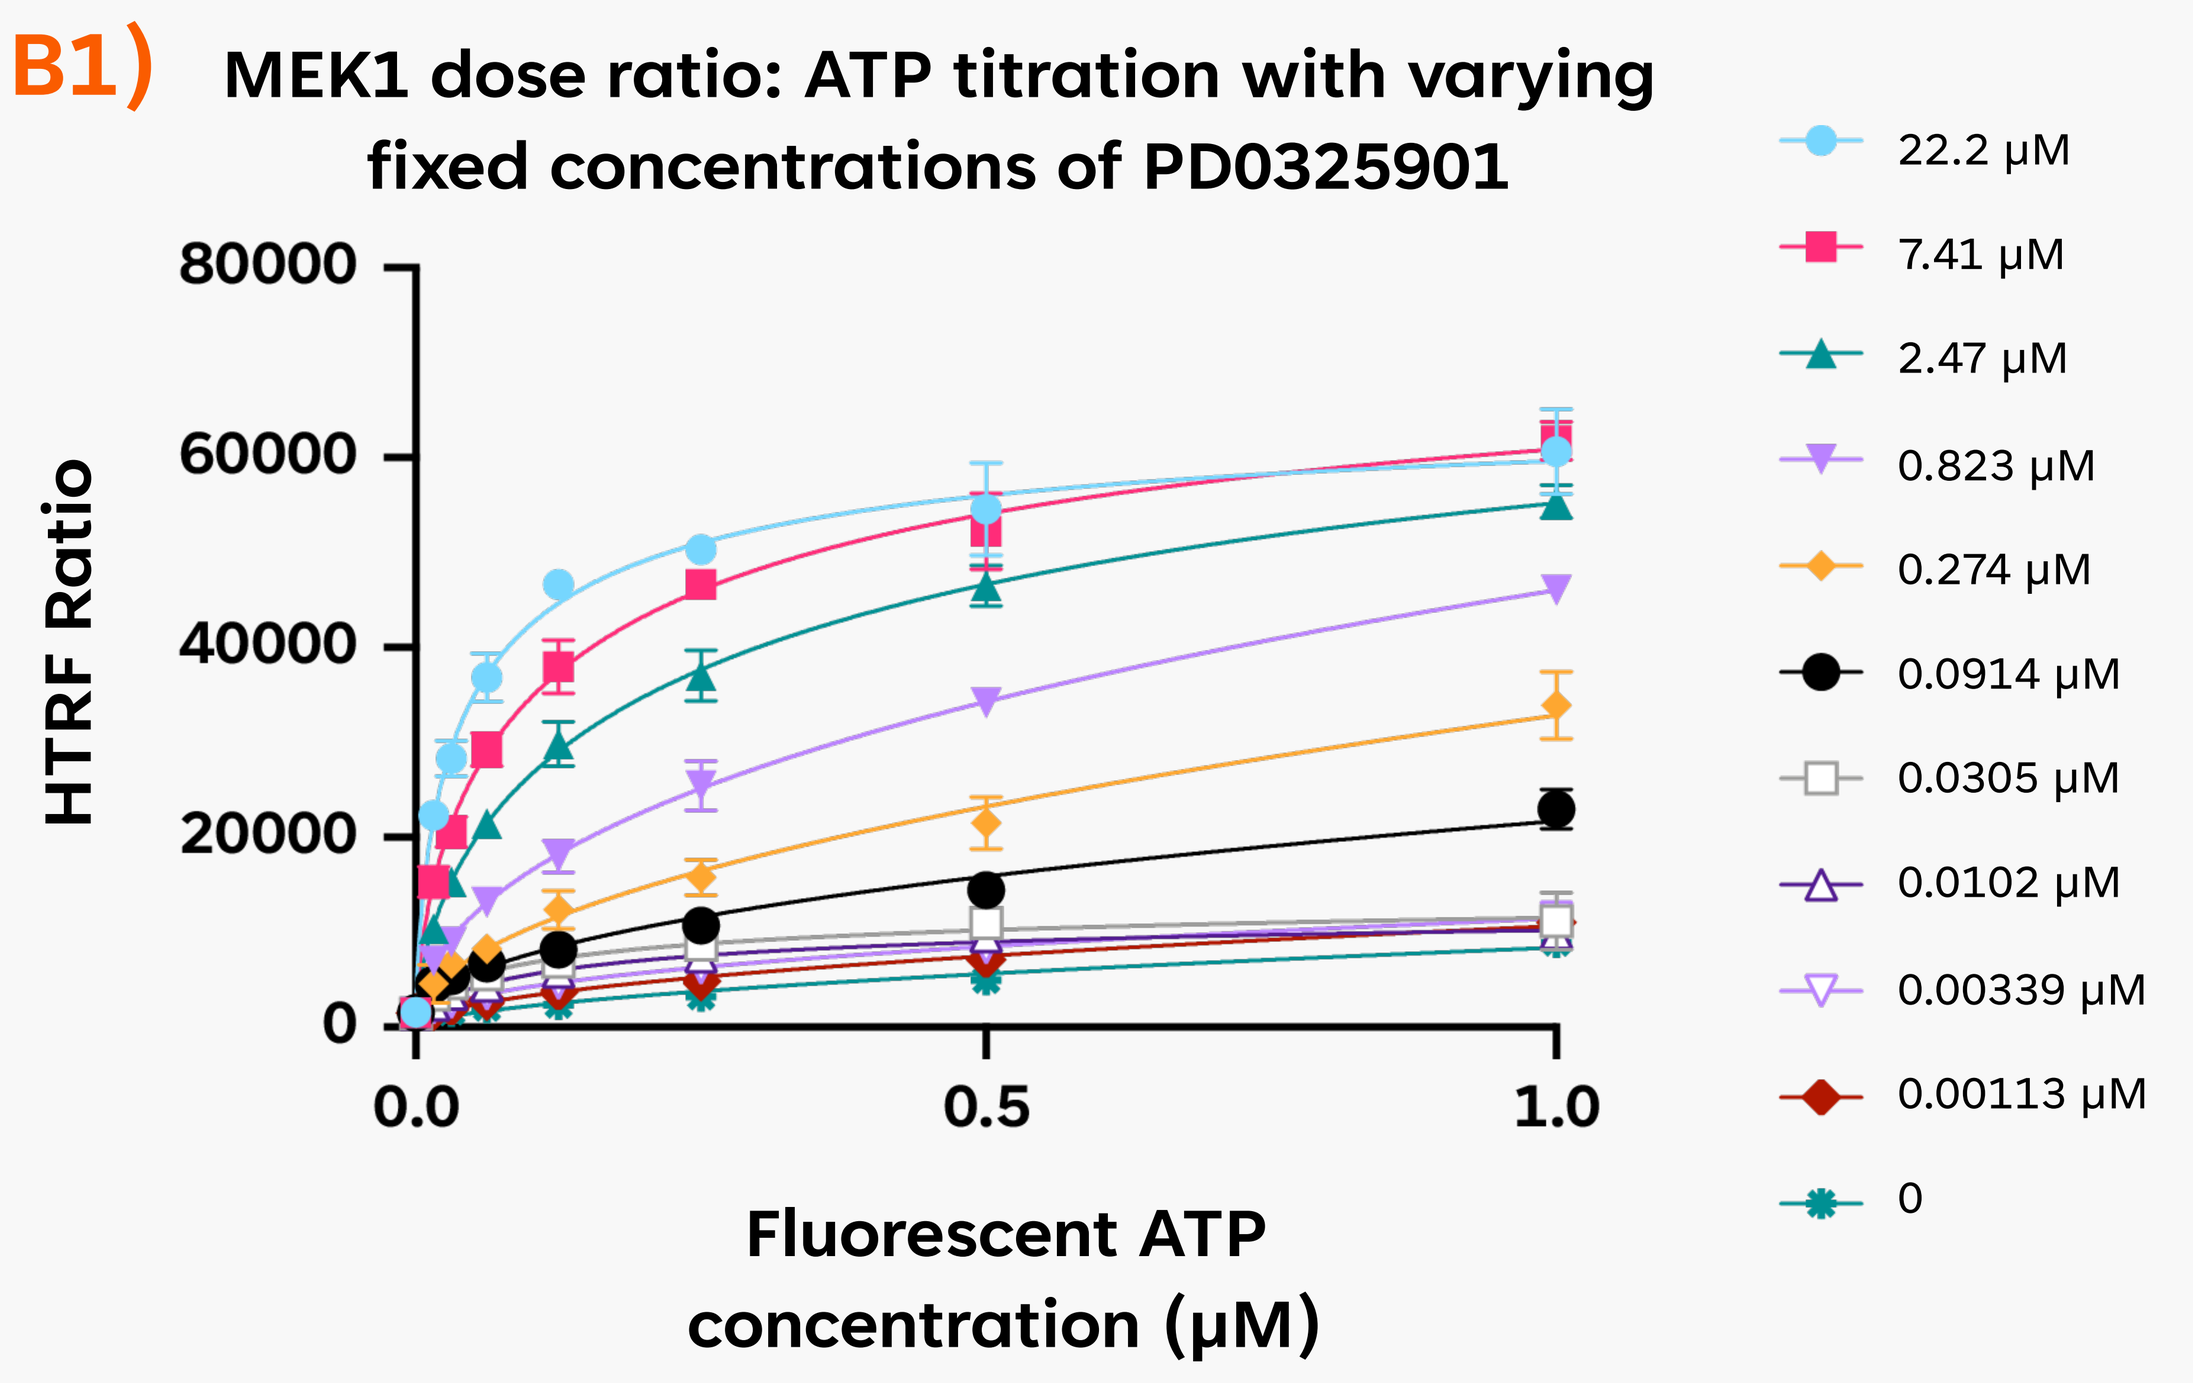
MEK1 dose ratio: ATP titration with varying fixed concentrations of PD0325901
HTRF Ratio
80000
60000
40000
20000
0
0.0
0.5
1.0
Fluorescent ATP concentration (μM)
22.2 μM
7.41 μM
2.47 μM
0.823 μM
0.274 μM
0.0914 μM
0.0305 μM
0.0102 μM
0.00339 μM
0.00113 μM
0

B2)

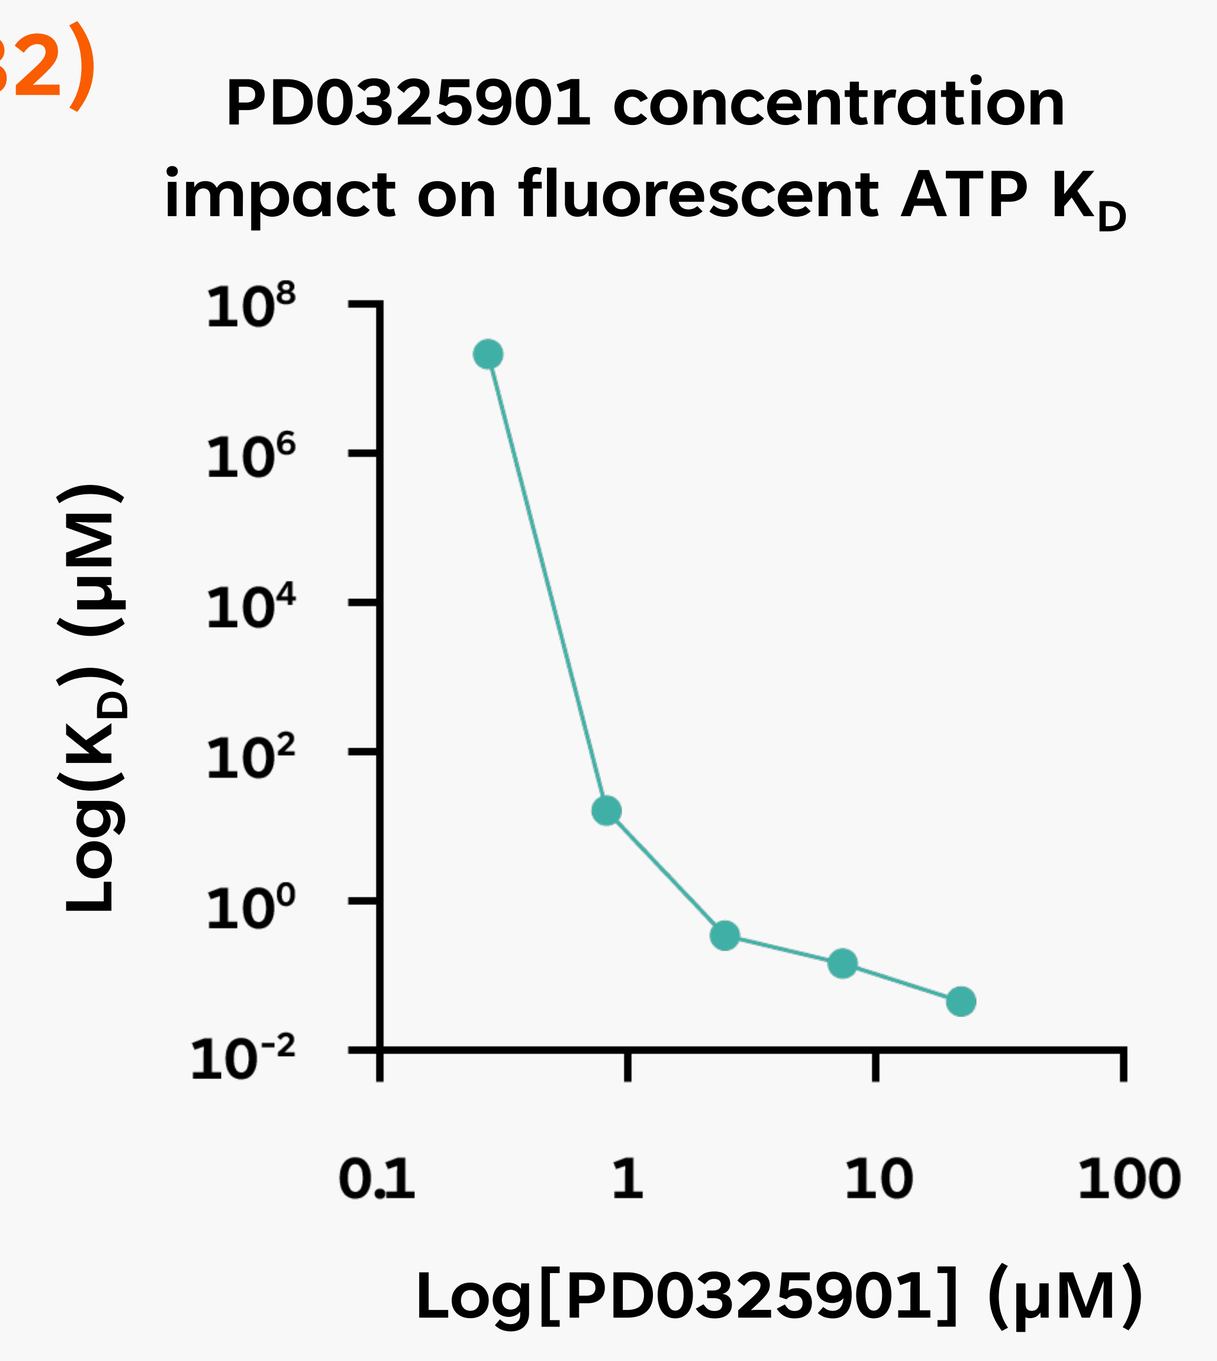
PD0325901 concentration impact on fluorescent ATP KD
Log(KD) (μM)
10^8
10^6
10^4
10^2
10^0
10^-2
0.1
1
10
100
Log[PD0325901] (μM)

Figure 2: (A1) Inhibitor EC_{50} shows a leftward shift, therefore increasing in potency as ATP concentration rises. **(A2)** Fluorescent ATP concentration impact on PD0325901 EC_{50}. **(B1)** Leftward shift in K_D is observed as concentration of inhibitor increases. **(B2)** PD0325901 concentration effect on fluorescent ATP K_D.

CONCLUSION

An inhibitor-dependent increase in assay signal was observed, due to more fluorescent ATP binding to MEK1 causing an increased FRET signal. The leftward shift in ATP K_D in the dose-ratio experiment, confirms that PD0325901 is promoting the binding of ATP, indicative of a type III inhibitor.

This proof-of-concept assay will be used to detect further type III kinase inhibitors, particularly for targets lacking reference compounds, and to potentially identify and characterise new groups of allosteric inhibitors.

Exscientia

For more information omedhurst@exscientia.co.uk