Cellular Target Engagement Approaches to Monitor Epigenetic Reader Domain Interactions.

A. Phillipou, C. Messenger, C. Carver, M. Mahmood, A. Lewis, P. Craggs and D. Brierley

GSK Medicines Research Centre, Gunnels Wood Road, Stevenage, Hertfordshire, SG1 2NY, UK

Introduction

Target engagement measures have traditionally been reliant on reductionist biochemical and biophysical techniques. However more recently technologies such as CETSA and BRET have allowed probing of target engagement in live cells. Promega's NanoBRET is a variation of BRET and consists fusion proteins with Nanoluc, a 19.1kDa luciferase which uses the substrate furimazine to produce high intensity bioluminescence and HaloTag a modified dehalogenase enzyme which can be selectivity labelled with a fluorophore. A key issue for clinical drug failure is safety concerns, recent studies show nearly 17% of clinical failures due to safety liability concerns[1]. BET proteins are key epigenetic regulators. Inhibition of BET proteins Brd4 and Brd2 shown to elicit an anti-inflammatory response in a wide range of therapeutic modalities along with cellular models such macrophages and T cells[2]. However, pan-BET inhibitors are not well tolerated[3]. Therefore, we utilised this NanoBRET PPI assay technology across the BET family, to drive both selectivity, ligand efficiency and offset potentially safety liability risks.

Assay technologies

3 high throughput protein::protein interactions (PPI) assays were developed in 384 well formats; utilising Promega's NanoBRET technology. BRD2, 3 and 4 Nanoluc fusion plasmids were generated alongside histone H3.3 Halotag fusion plasmids and transiently transfected into HEK 293 cells. Disruption of BRD binding to H3.3 was monitored by a reduction in BRET signal.
Alongside the PPI assays a BRD4 NanoBRET tracer assay was also developed using a bodipy labelled bromosporine as a tracer, competitive displacement of this tracer reduces BRET between the tracer and BRD-Nluc fusion.

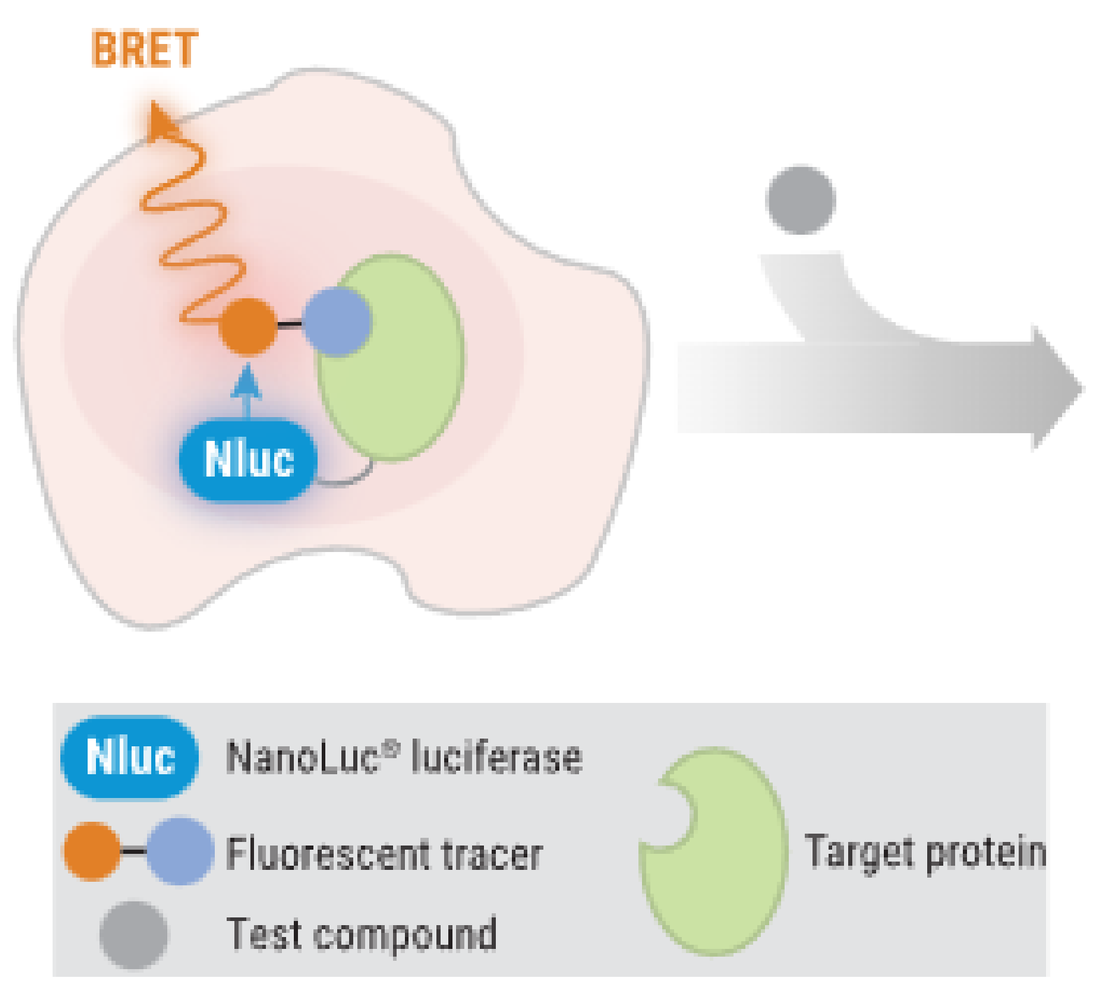
BRET
Nluc
Nluc
Nluc NanoLuc® luciferase
Fluorescent tracer
Test compound
Target protein

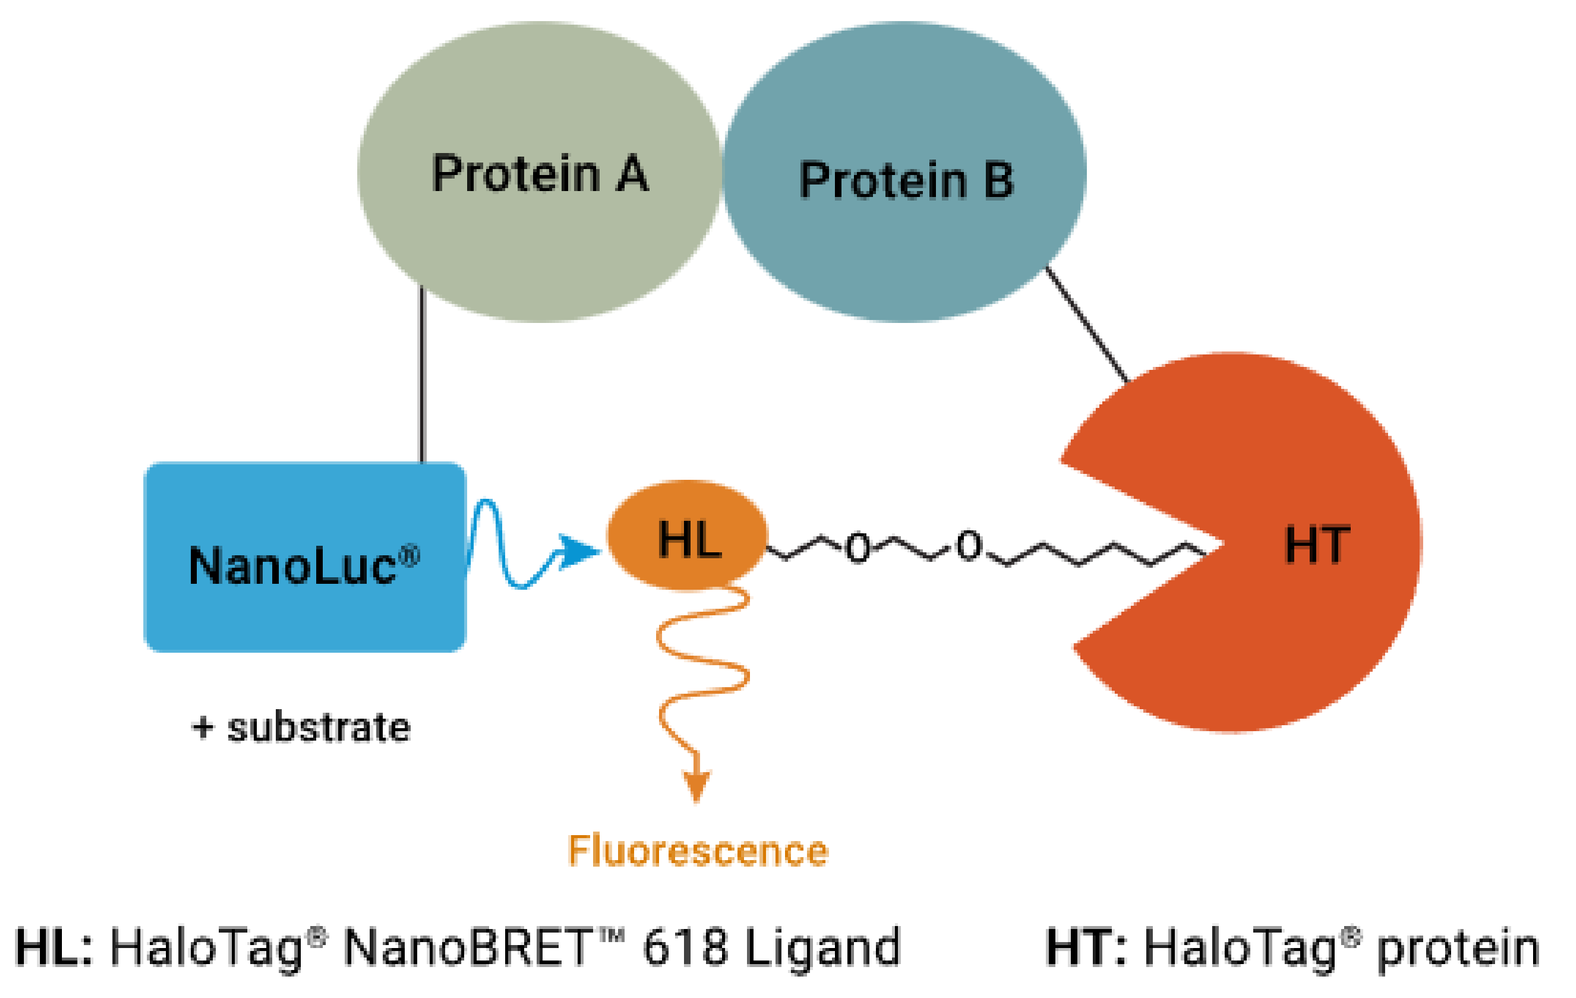
Protein A
Protein B
NanoLuc®
+ substrate
HL
HT
Fluorescence
HL: HaloTag® NanoBRET™ 618 Ligand
HT: HaloTag® protein

Results

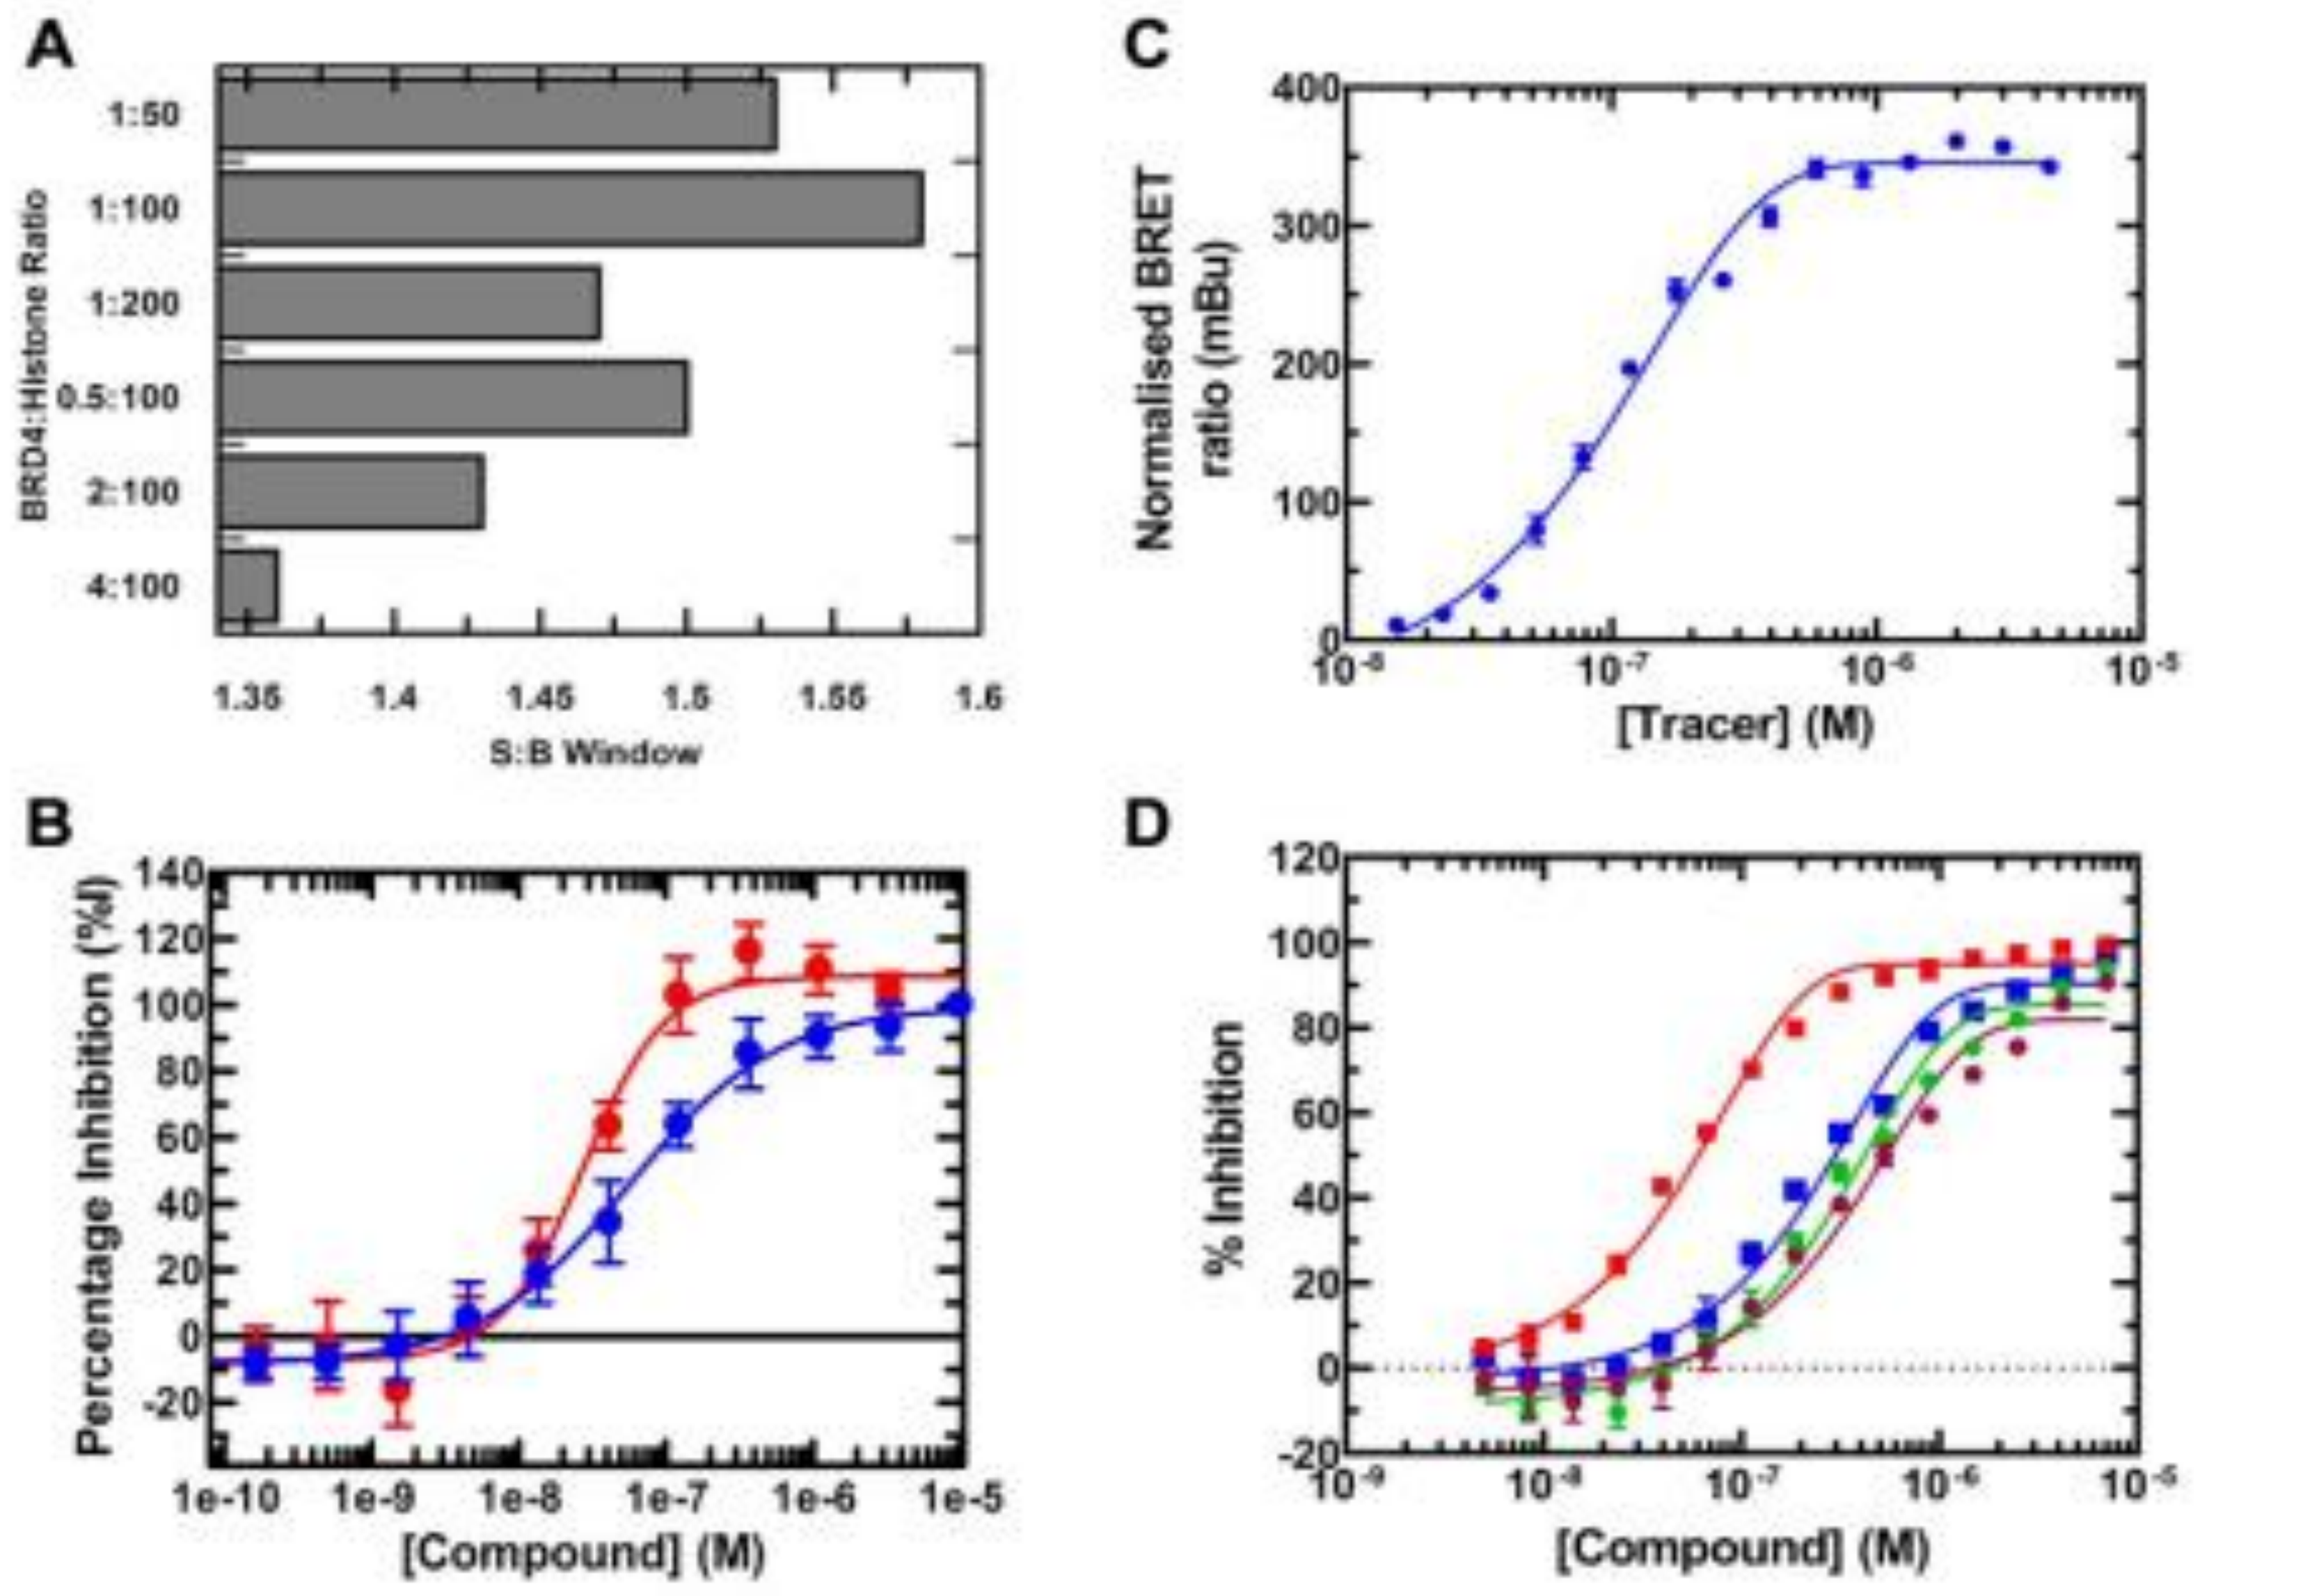
A
BRD4:Histone Ratio
S:B Window
B
Percentage Inhibition (%I)
[Compound] (M)
C
Normalised BRET ratio (mBu)
[Tracer] (M)
D
% Inhibition
[Compound] (M)

Fig. 1 (A) variations in S:B driven by differing donor and acceptor ratios in Brd4 PPI assay. (B) I-BET151 (•) and (+)-JQ1 (•) dose response curves in the Brd4 PPI assay. (C) initial BSP-BODIPY tracer characterisation to maximise signal window. (D) PFI-1 (•), bromosporine (•), I-BET151 (■), and (+)-JQ1 (■) dose response curves in the Brd4 tracer assay.

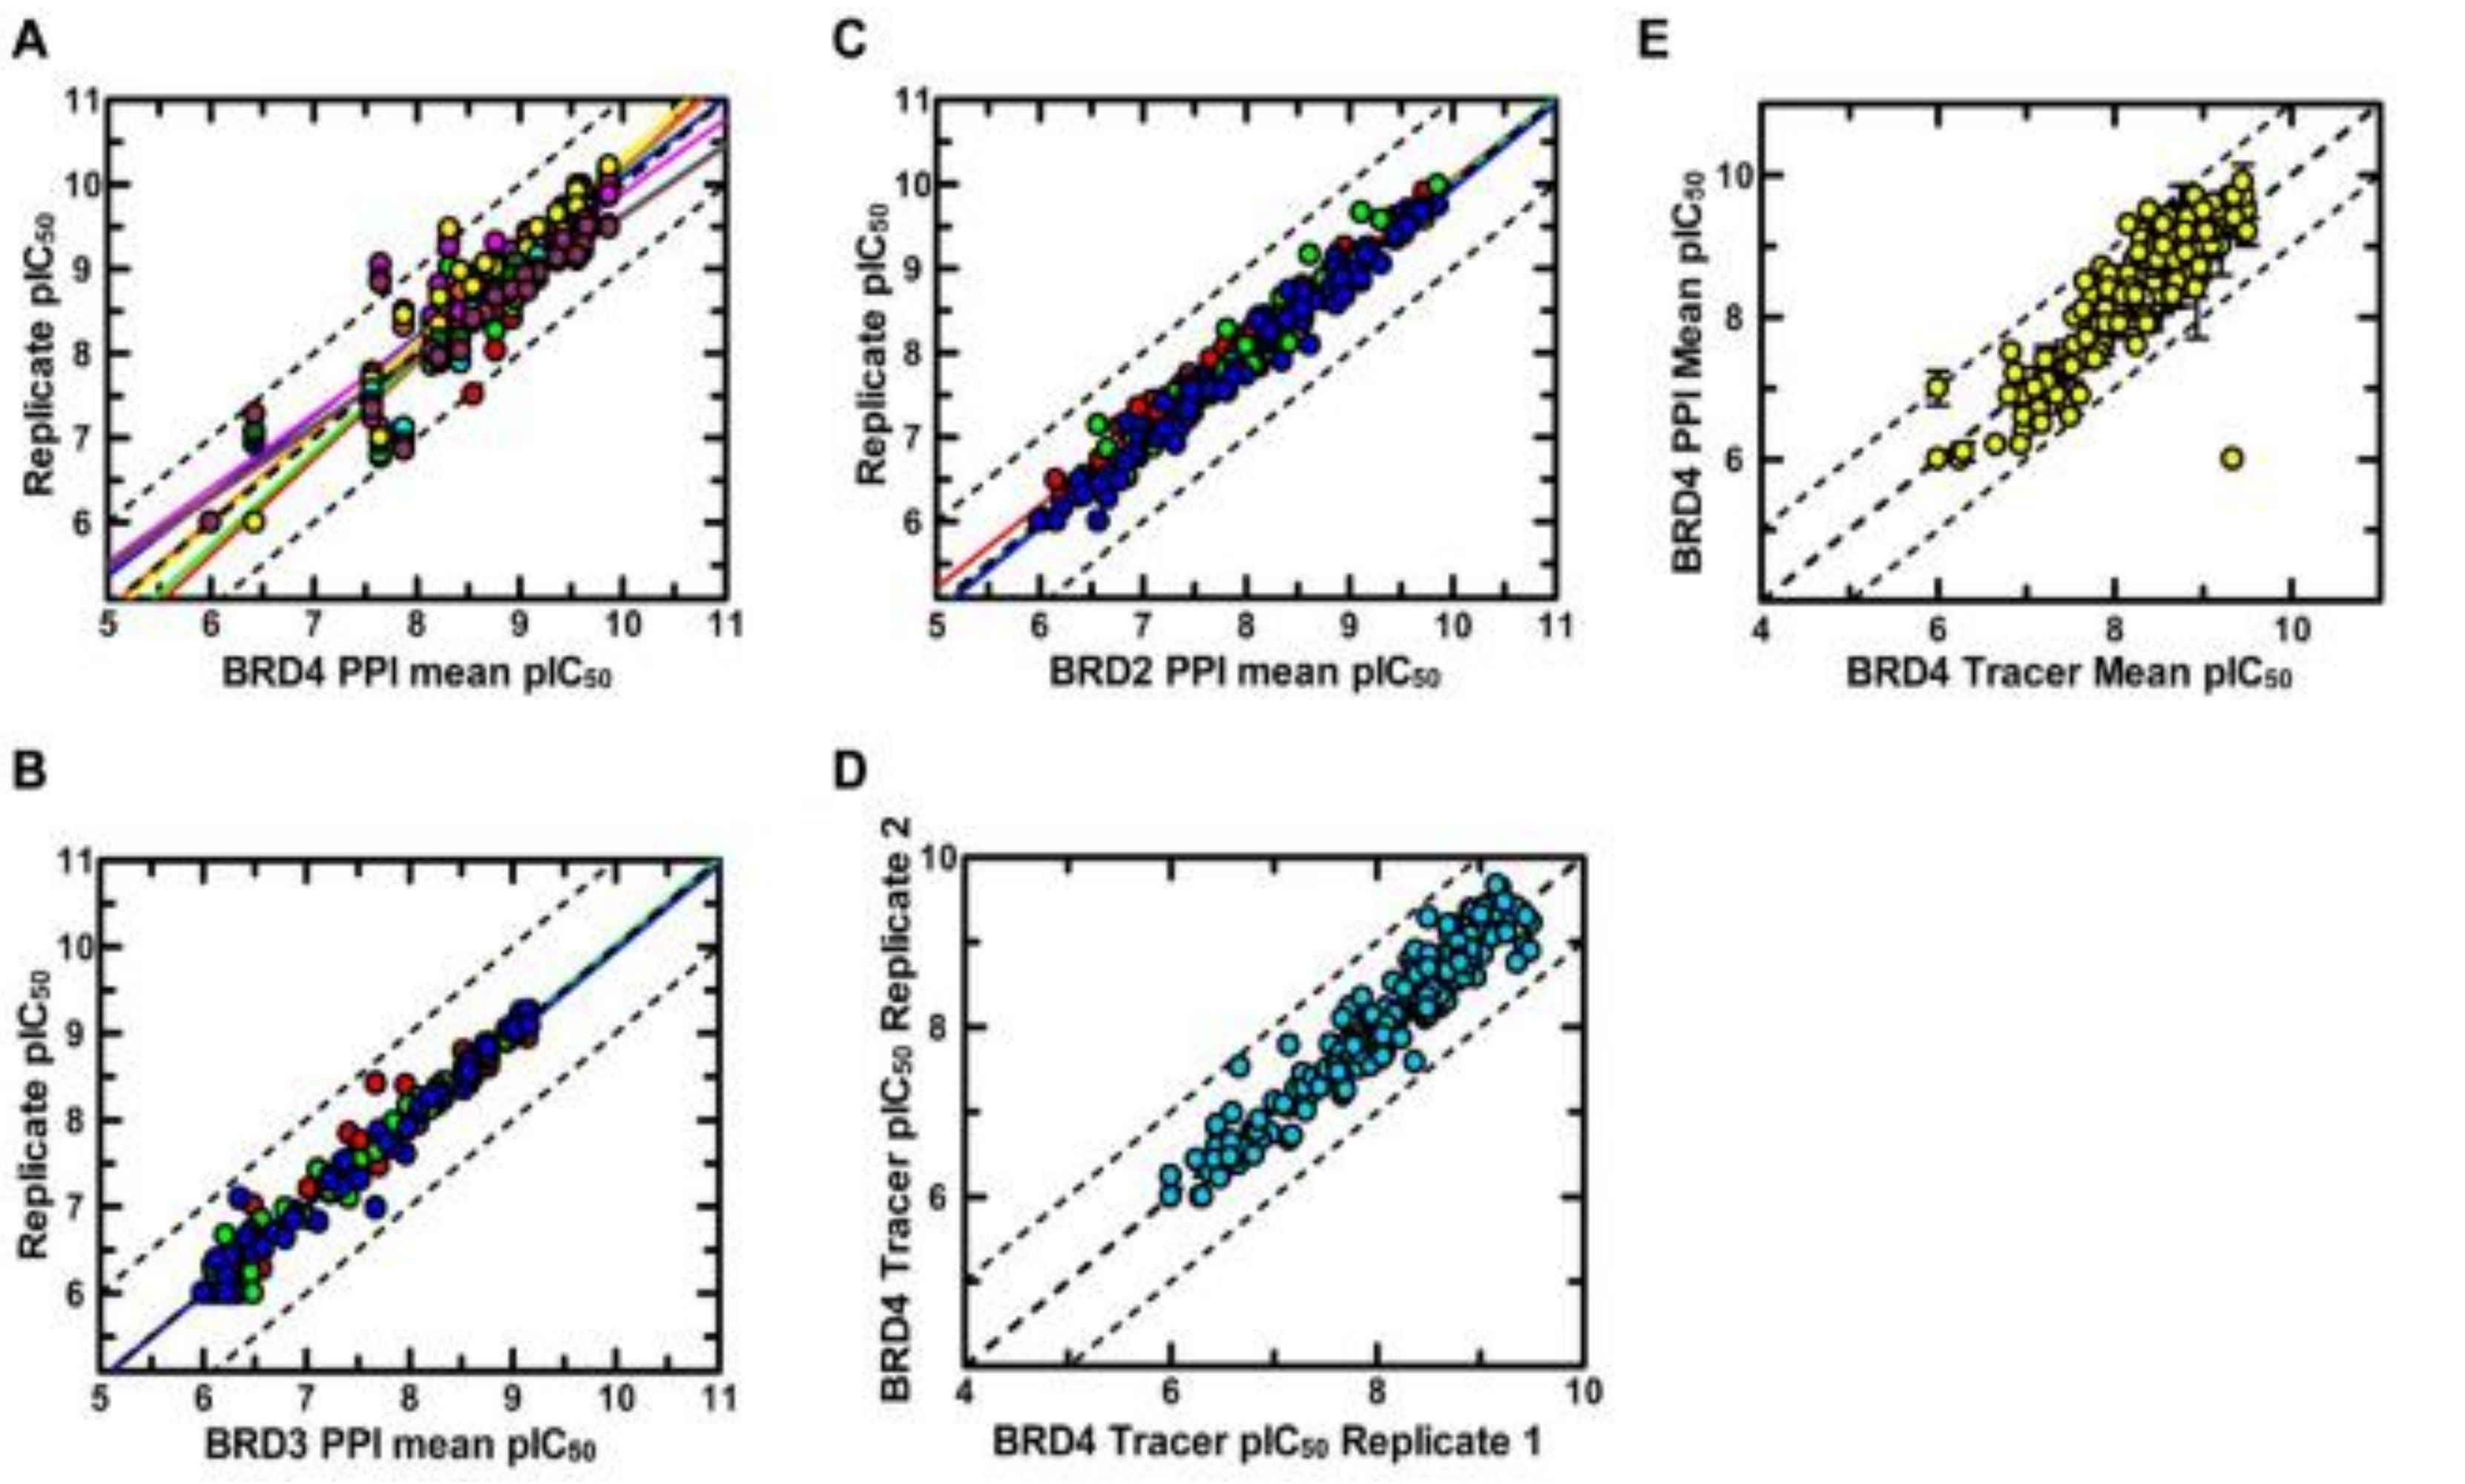
A
Replicate pIC50
BRD4 PPI mean pIC50
C
Replicate pIC50
BRD2 PPI mean pIC50
E
BRD4 PPI Mean pIC50
BRD4 Tracer Mean pIC50
B
Replicate pIC50
BRD3 PPI mean pIC50
D
BRD4 Tracer pIC50 Replicate 2
BRD4 Tracer pIC50 Replicate 1

Fig.2 Diverse set of BET inhibitors profiled across NanoBRET PPI and tracers assay. Strong correlation between test occasions for all PPI assays, A r^2=0.79, B and C r^2 =0.99.Differing colours on plot refer to discrete test occasions. Strong correlation between Brd4 tracer and Brd4 PPI assays.

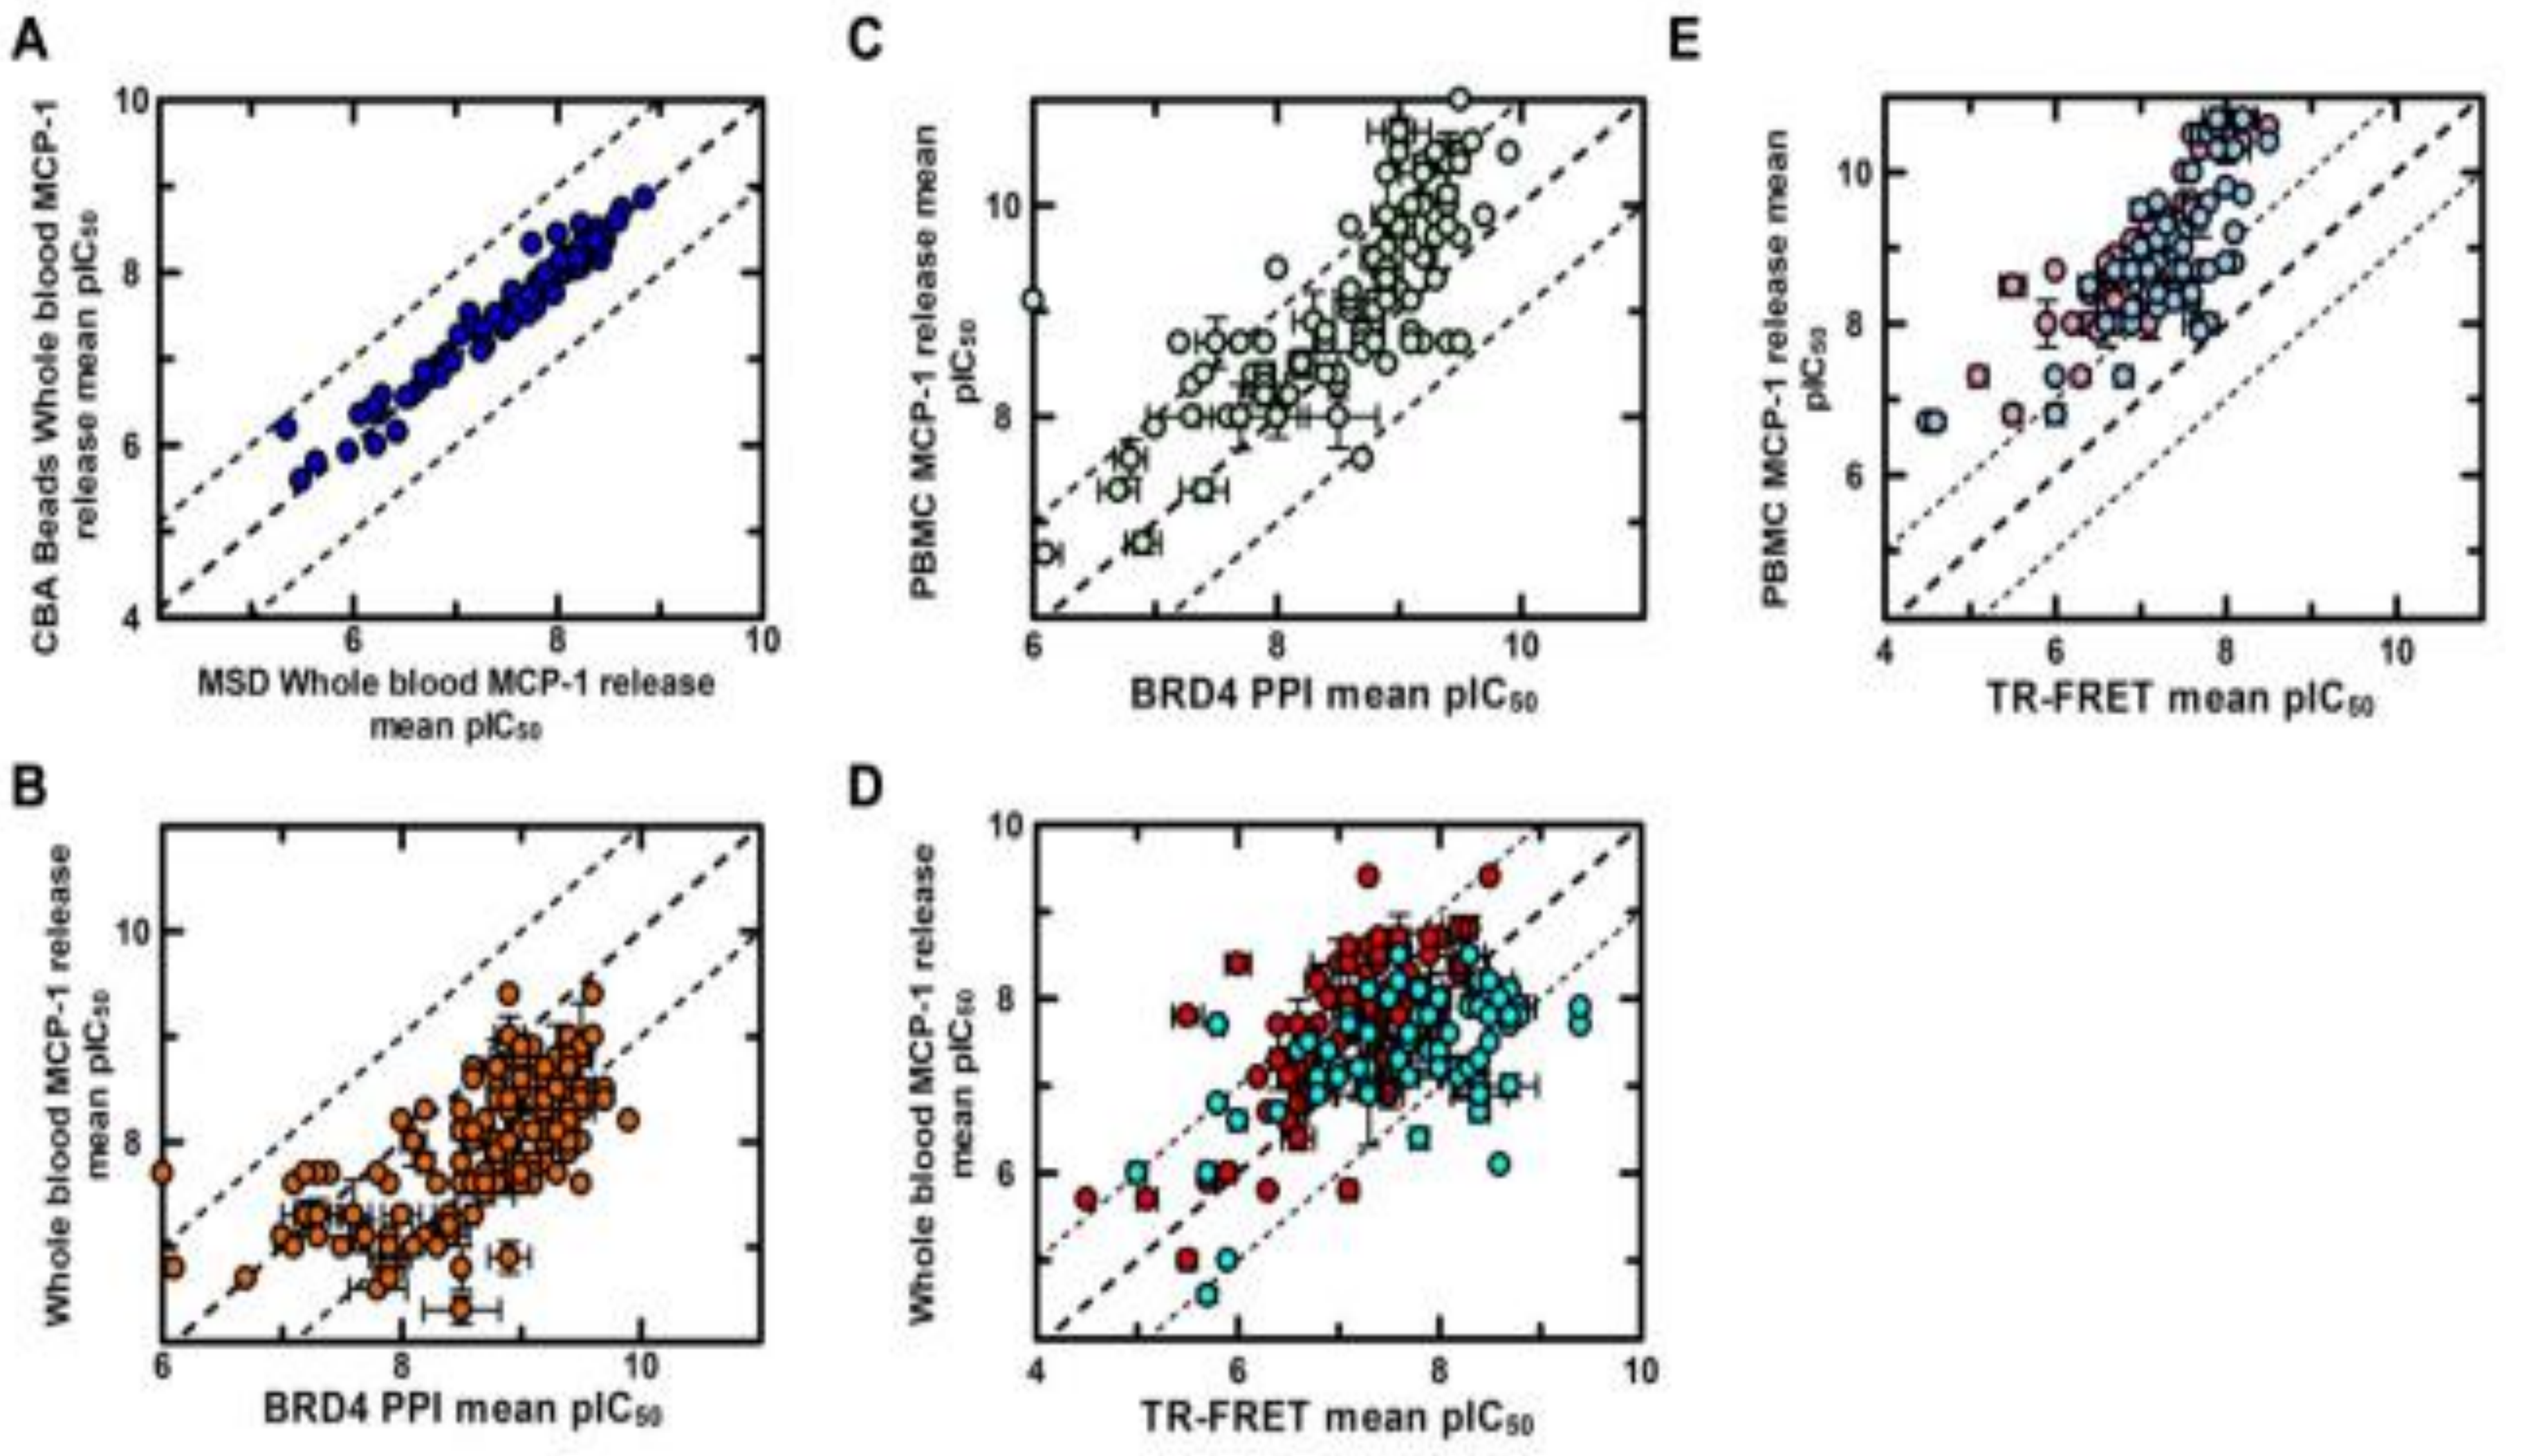
A
CBA Beads Whole blood MCP-1 release mean pIC50
MSD Whole blood MCP-1 release mean pIC50
C
PBMC MCP-1 release mean pIC50
BRD4 PPI mean pIC50
E
PBMC MCP-1 release mean pIC50
TR-FRET mean pIC50
B
Whole blood MCP-1 release mean pIC50
BRD4 PPI mean pIC50
D
Whole blood MCP-1 release mean pIC50
TR-FRET mean pIC50

Fig. 3 Two cytokine release assays were developed historically, LPS stimulated hWB and PBMC models with MCP-1 endpoints. (B) and (C) demonstrate a clear link between efficacy of cellular TE and phenotypic outcome, r^2=0.56 for both. (D) and (E) BRD4 TR-FRET data was also correlated to the phenotypic data, ranked order was conserved.

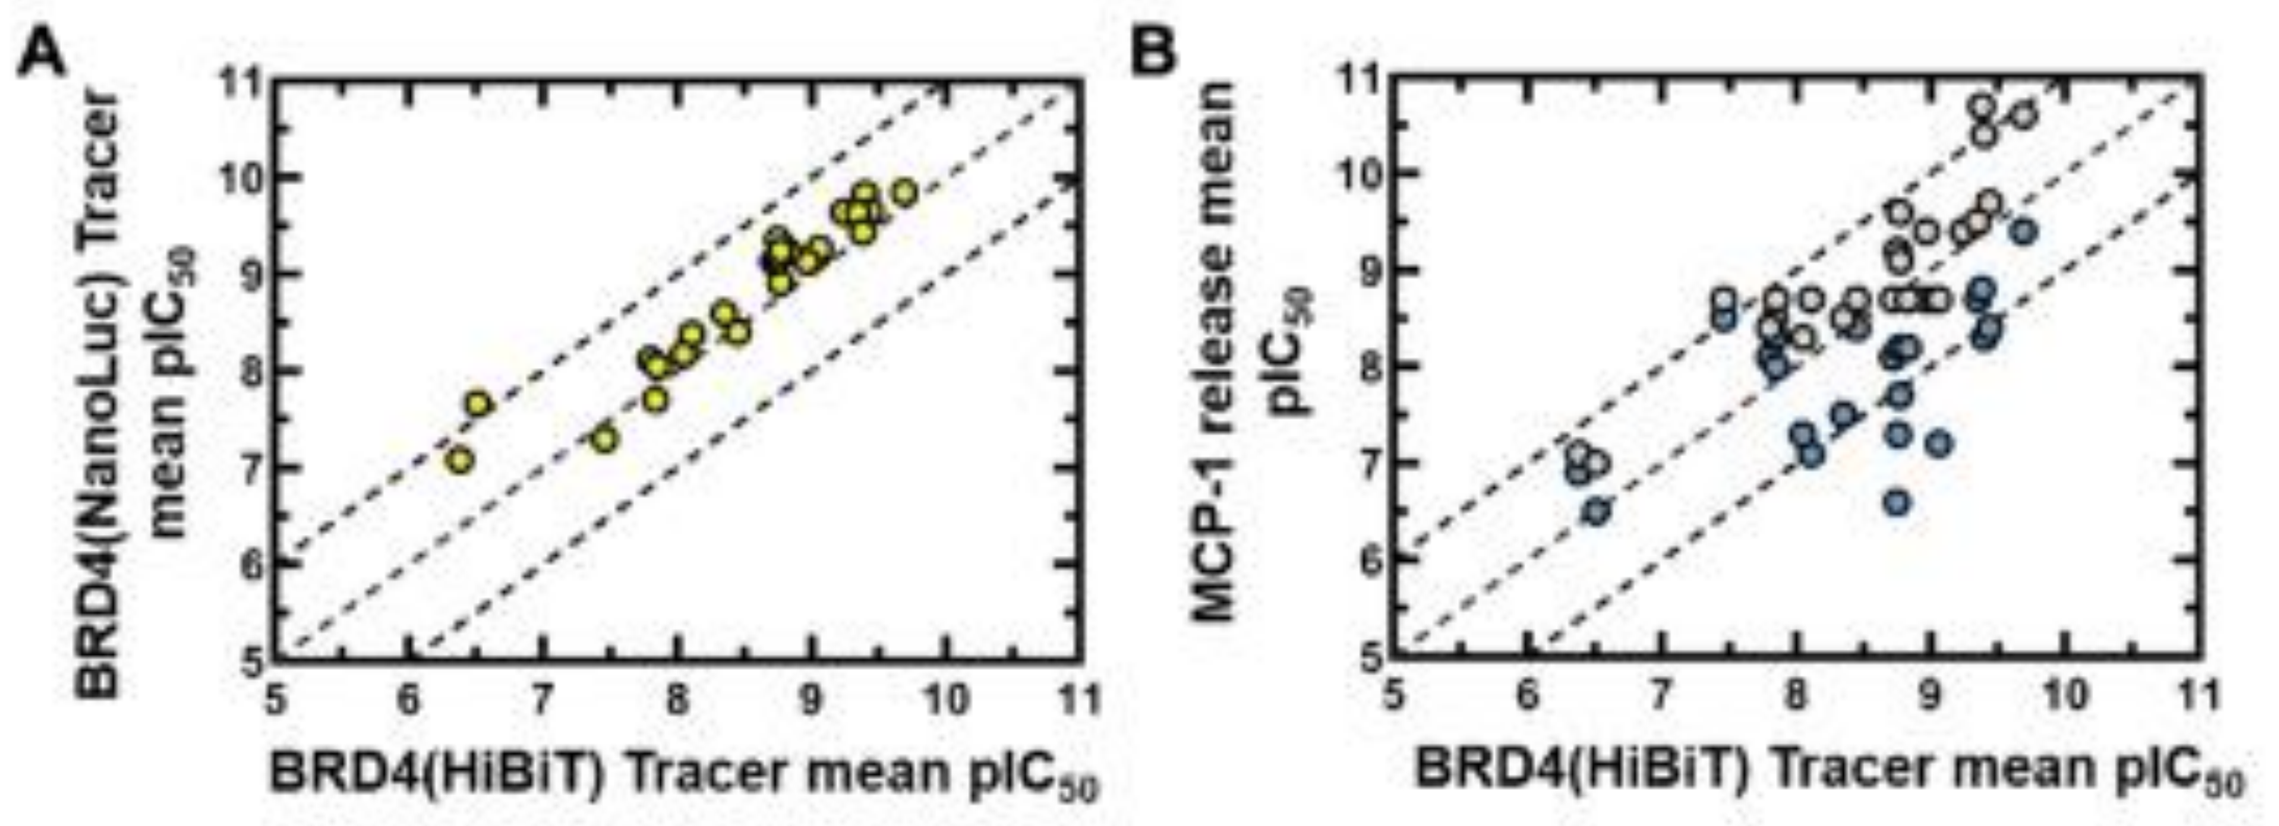
A
BRD4(NanoLuc) Tracer mean pIC50
BRD4(HiBiT) Tracer mean pIC50
B
MCP-1 release mean pIC50
BRD4(HiBiT) Tracer mean pIC50

Fig. 4 HiBiT tag is an 11 AA peptide that binds LgBiT (Kd=0.7nM) to form a functional luciferase. HiBiT tagging of endogenous protein gives a more consistent expression of HiBiT-Brd4 in contrast to the Nluc-Brd4 overexpression assay system. (A) Brd4 HiBiT tracer assay correlated well to the Nluc-BRD4 tracer assay r^2=0.90. (B) Interestingly the HiBiT assay correlated better to the phenotypic assay r^2=0.76 than the transient NanoBRET assay systems.

Conclusion

- The HT-Cellular target engagement PPI assays have been shown to be robust and reliable with r^2>0.9 across all isoforms developed.
- These assays can be embedded within the early drug discovery screening cascades to dial in ligand selectivity.
- These assays have the potential to bridge the gap between lower throughput phenotypic assays and more reductionist ligand::protein biochemical models.

The human biological samples were sourced ethically and their research use was in accord with the terms of the informed consents under an IRB/EC approved protocol.

References
1. Hwang TJ, Carpenter D, Lauffenburger JC, Wang B, Franklin JM, Kesselheim AS. Failure of Investigational Drugs in Late-Stage Clinical Development and Publication of Trial Results. JAMA Intern Med. 2016;176(12):1826–1833. doi:10.1001/jamainternmed.2016.6008
2. Nicodeme, E., Jeffrey, K., Schaefer, U. *et al.* Suppression of inflammation by a synthetic histone mimic. *Nature* **468**, 1119–1123 (2010). https://doi.org/10.1038/nature09589.
3. Sandy Amorim, Anastasios Stathis, Mary Gleeson, Sunil Iyengar, Valeria Magarotto, Xavier Leleu, Franck Morschhauser, Lionel Karlin, Florence Broussais, Keyvan Rezai, Patrice Herait, Carmen Kahatt, François Lokiec, Gilles Salles, Thierry Facon, Antonio Palumbo, David Cunningham, Emanuele Zucca, Catherine Thieblemont, Bromodomain inhibitor OTX015 in patients with lymphoma or multiple myeloma: a dose-escalation, open-label, pharmacokinetic, phase 1 study, The Lancet Haematology, Volume 3, Issue 4,2016, Pages e196-e204,ISSN 2352-3026,https://doi.org/10.1016/S2352-3026(16)00021-1.

ELRIG, Drug Discovery, 2022